CCL24 BLOCKADE ATTENUATES BILIARY INFLAMMATION BY INTERFERING WITH MONOCYTE AND NEUTROPHIL RECRUITMENT

Chemomab THERAPEUTICS

Raanan Greenman[1], Ilan Vaknin[1], Michal Segal-Salto[1], Avi Katav[1], Omer Levy[1], Ophir Hay[2], Inbal Mishalian[2], Devorah Olam[2], Amnon Peled[2], Adi Mor[1]

1 Chemomab Ltd., Israel,

2 Goldyne Savad Institute of Gene Therapy, Hebrew University Hospital, Jerusalem, Israel

Introduction

Primary Sclerosing Cholangitis (PSC) is characterized by the damaged peribiliary space, in which immune cells, reactive bile epithelial cells and fibroblasts from different sources interact, leading to elevation of cytokines, chemokines and a variety of other secreted factors that contribute to the inflammatory process. The chemokine CCL24 has been shown to help drive these self-reinforcing processes in fibrotic-inflammatory diseases, and its blockade was shown to ameliorate PSC progression in animal models.

Aim

- We aim to study the role of CCL24, a chemokine that promotes immune cell trafficking and activation as well as pro-fibrotic activities at inflammatory sites in PSC pathophysiology.
- Specifically, we examined the role of CCL24 and its blockade using CM-101, a CCL24 neutralizing antibody, on the trafficking of immune cells to tissues with a high presence of CCL24.

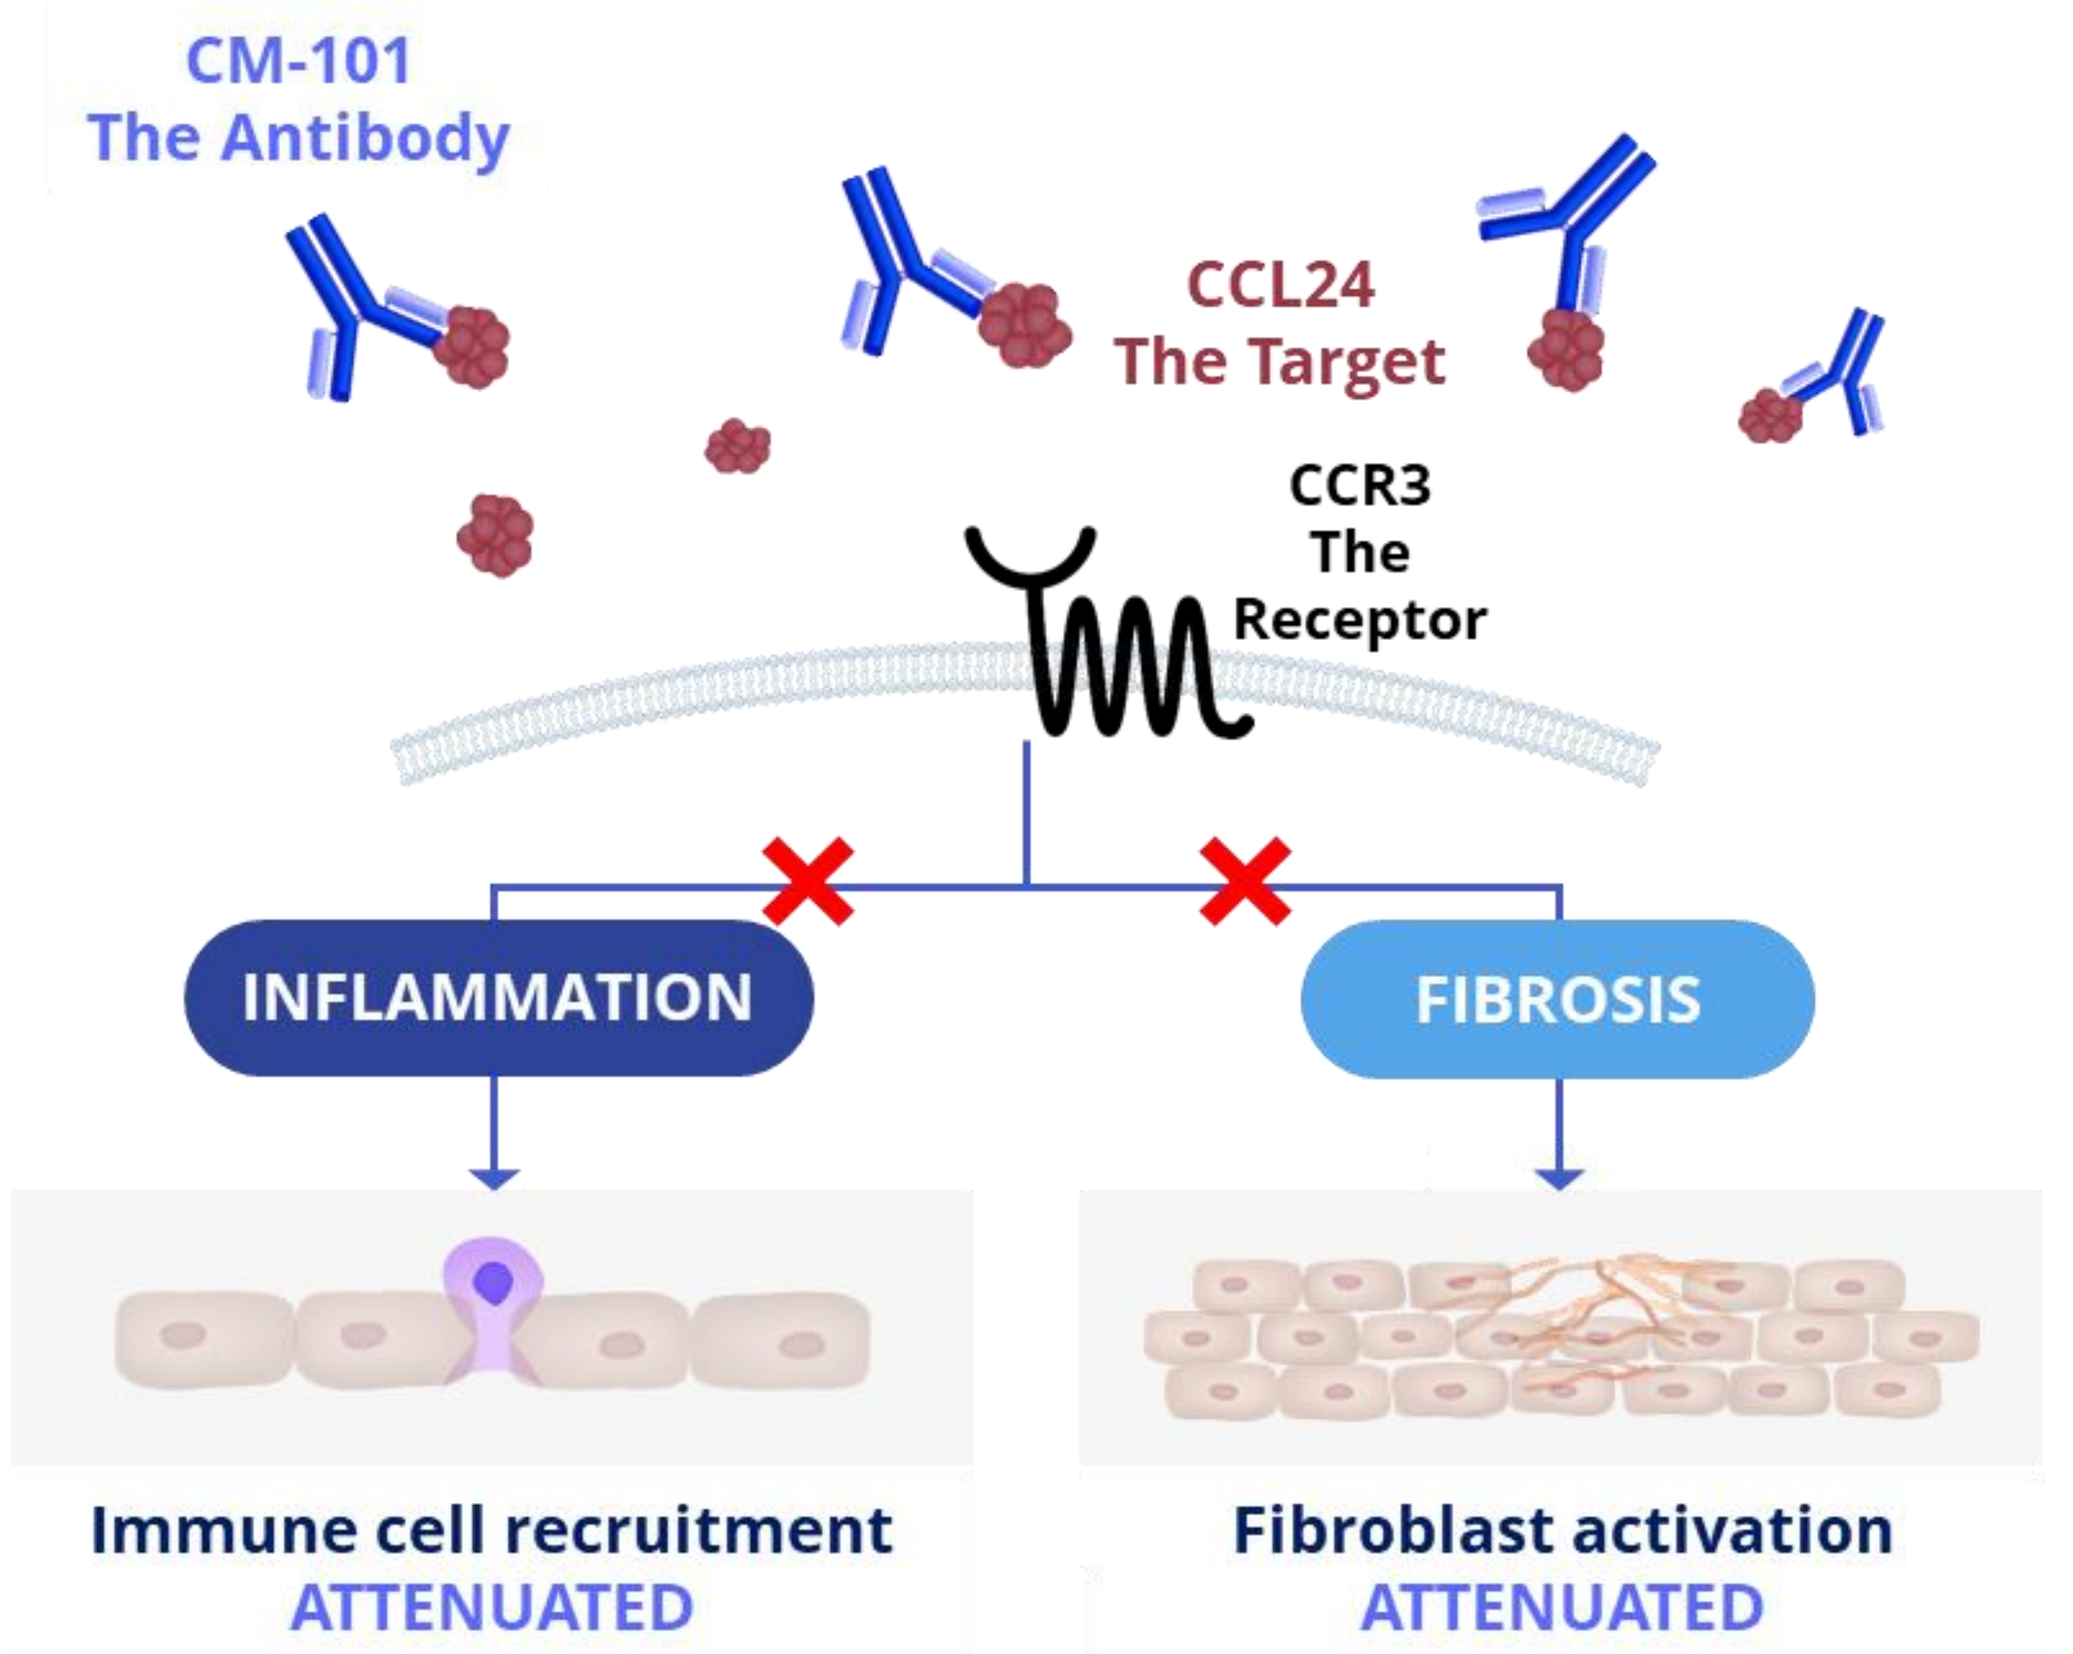

CM-101
The Antibody
CCL24
The Target
CCR3
The
Receptor
INFLAMMATION
FIBROSIS
Immune cell recruitment
ATTENUATED
Fibroblast activation
ATTENUATED

Methods

- Immune cell recruitment to CCL24-enriched areas was examined in diseased liver and following intraperitoneal (i.p.) injection of CCL24.
- We treated Mdr2-/- mice, a PSC model, with the neutralizing antibody CM-101. Immune cell composition was analyzed in blood and liver by FACS and immunohistochemistry.
- Peritoneal immune cell populations of intraperitoneally-injected animals were then analyzed by single-cell RNA sequencing (scRNA-seq) and by FACS.

CM-101 Treatment in PSC Mouse Model

- CCL24 blockade using CM-101 inhibited accumulation of peribiliary macrophages and neutrophils.
- These changes in immune cell populations highly correlate with disease severity, marked by collagen disposition, biliary mass and serum ALP levels.

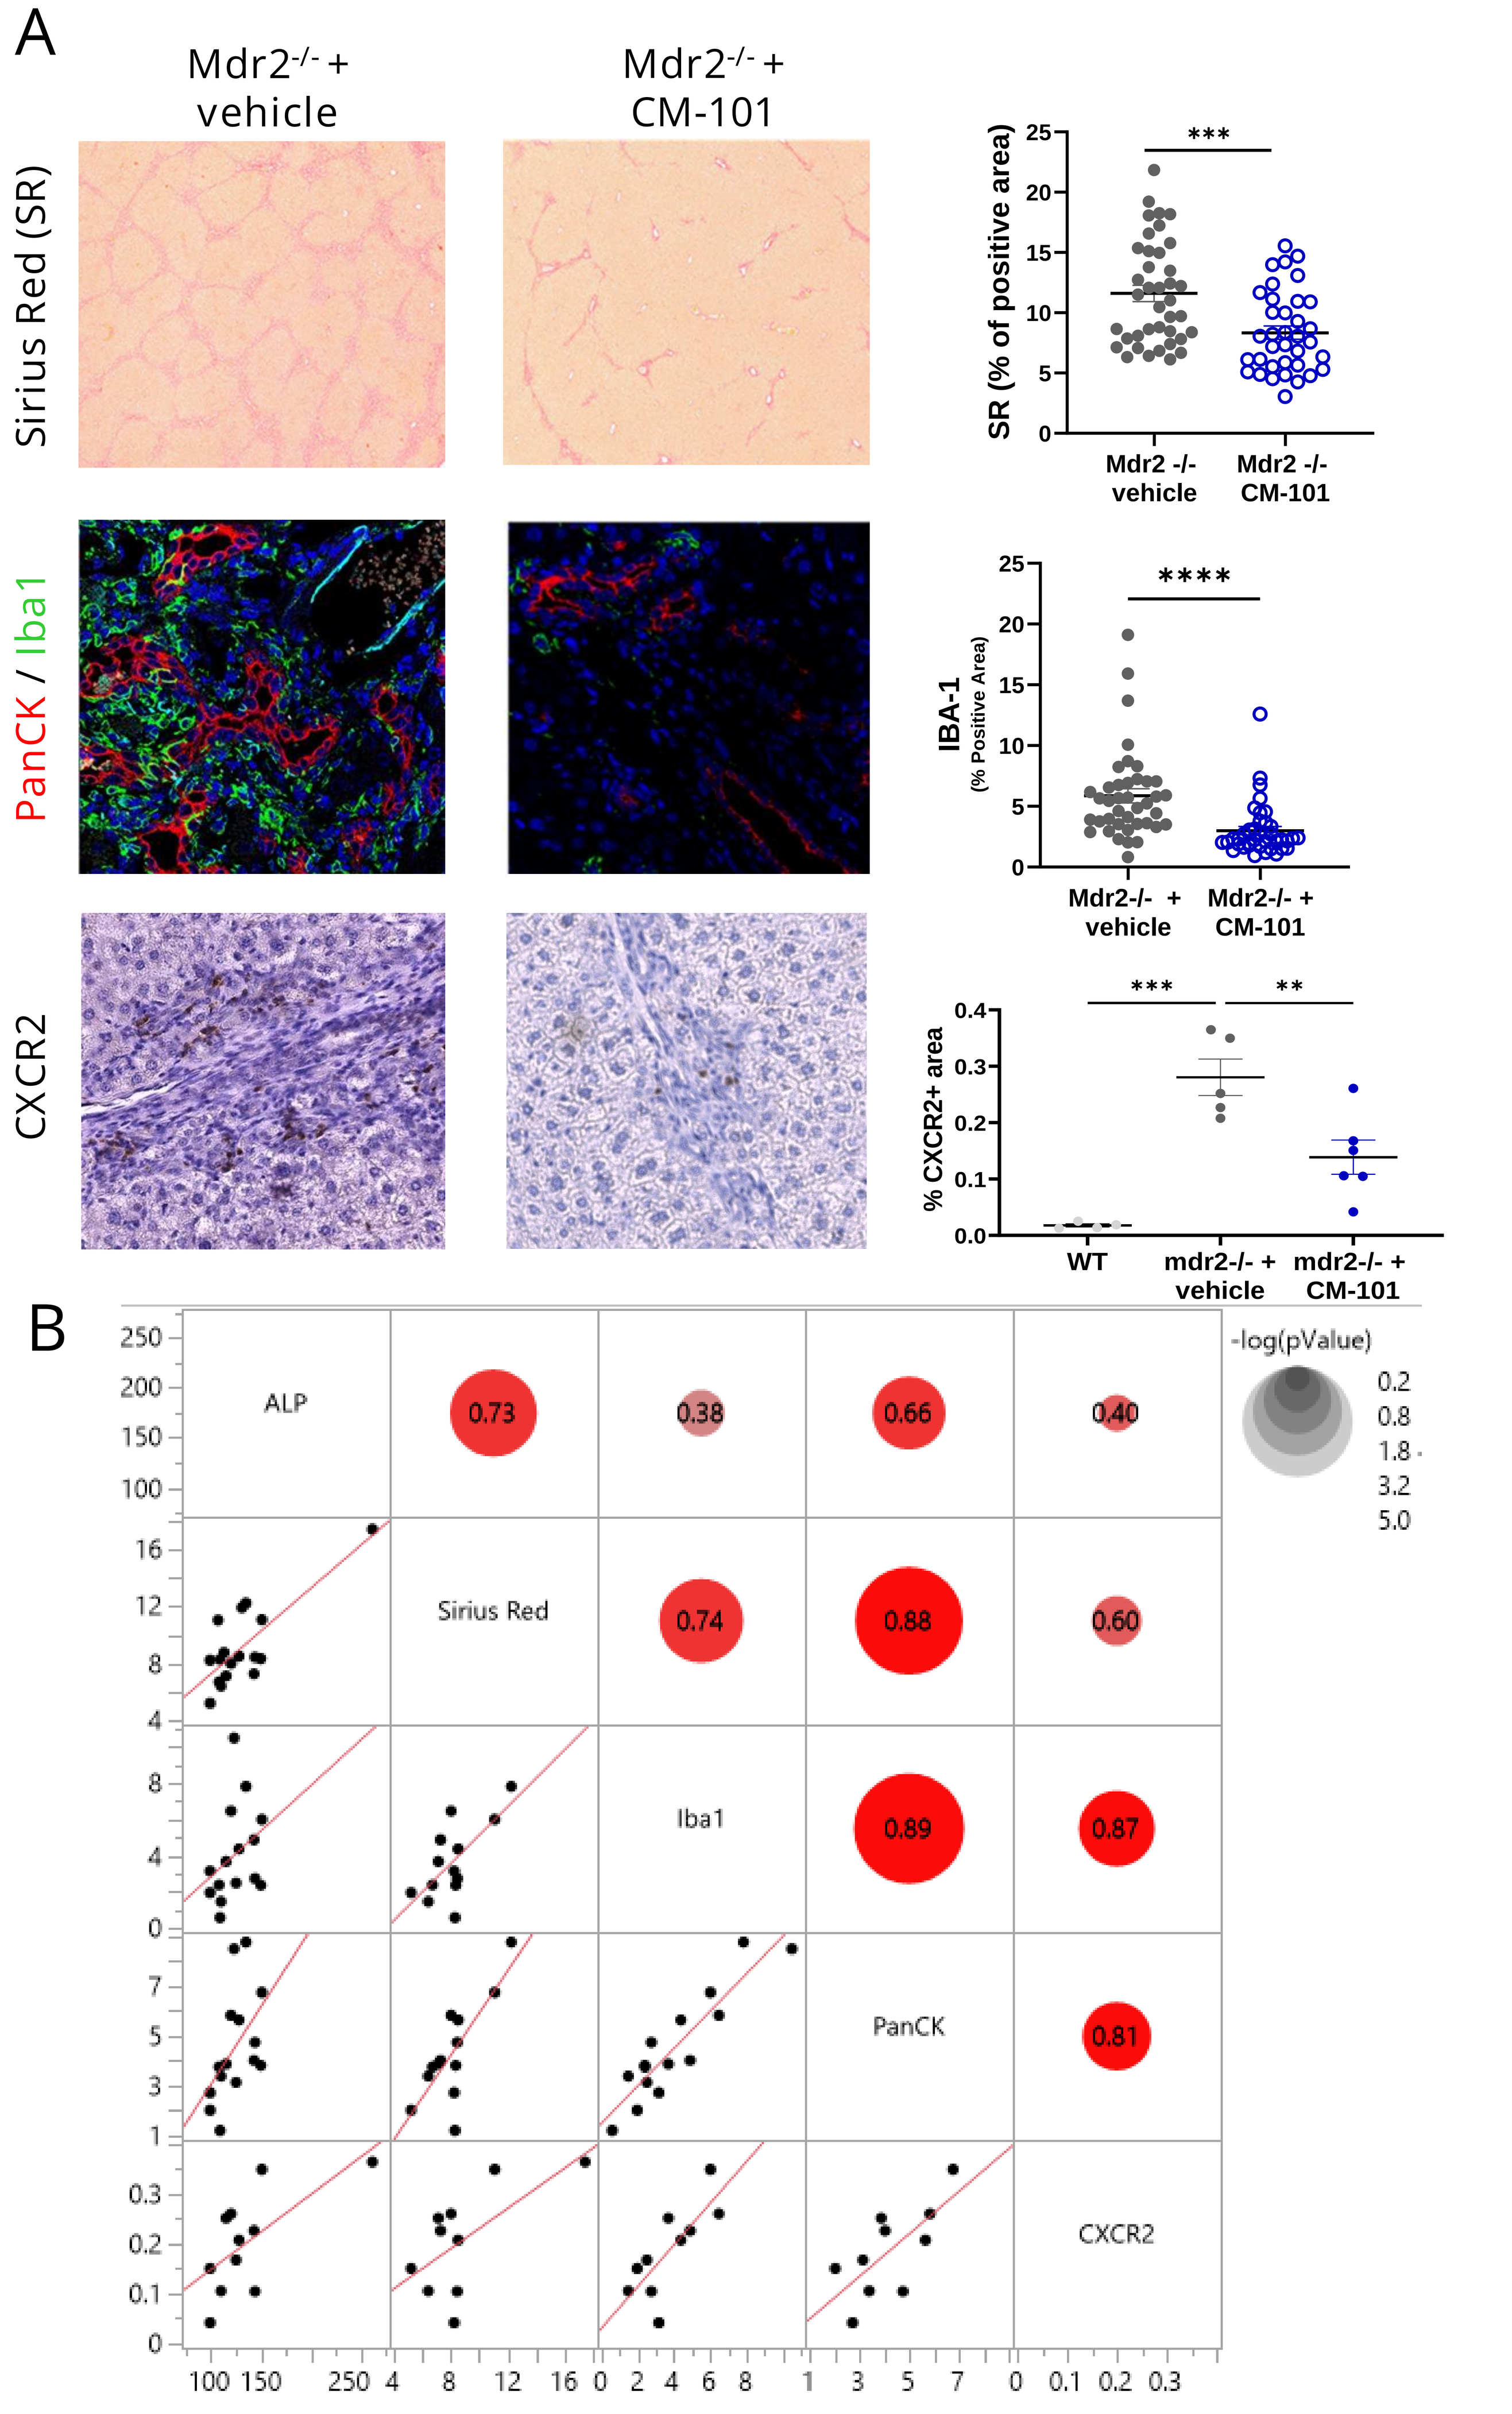

A
Mdr2-/- + vehicle
Mdr2-/- + CM-101
Sirius Red (SR)
PanCK / Iba1
CXCR2
SR (% of positive area)
Mdr2 -/- vehicle
Mdr2 -/- CM-101
IBA-1 (% Positive Area)
Mdr2-/- + vehicle
Mdr2-/- + CM-101
% CXCR2+ area
WT
mdr2-/- + vehicle
mdr2-/- + CM-101
B
ALP
Sirius Red
Iba1
PanCK
CXCR2
0.73
0.38
0.66
0.40
0.74
0.88
0.60
0.89
0.87
0.81
-log(pValue)

Mdr2-/- mice treated with CM-101 exhibit reduced liver damage, fibrosis and inflammation. (A) liver staining for fibrosis (Sirius Red), cholangiocyte (PanCK), macrophage (Iba1) and neutrophil (CXCR2) accumulation. Left panels, representative images; right panels, quantification ± SEM. (B) Correlation matrix between serum ALP levels, Sirius Red positive stain area, Iba1 positive stain area, PanCK positive stain area and CXCR2 positive stain area. Right side of the matrix presents Pearson's correlation (r) for each pair. p Value is visualized by circle's size.

CCL24 i.p. Injection

- CCL24 i.p. injection induced recruitment of monocytes and neutrophils, in a manner distinctive to CCL24 (compared with CCL11 injection), and upregulated CCL24 expression in macrophages and dendritic cells.
- CM-101 pretreatment prevented recruitment of monocytes and neutrophils.

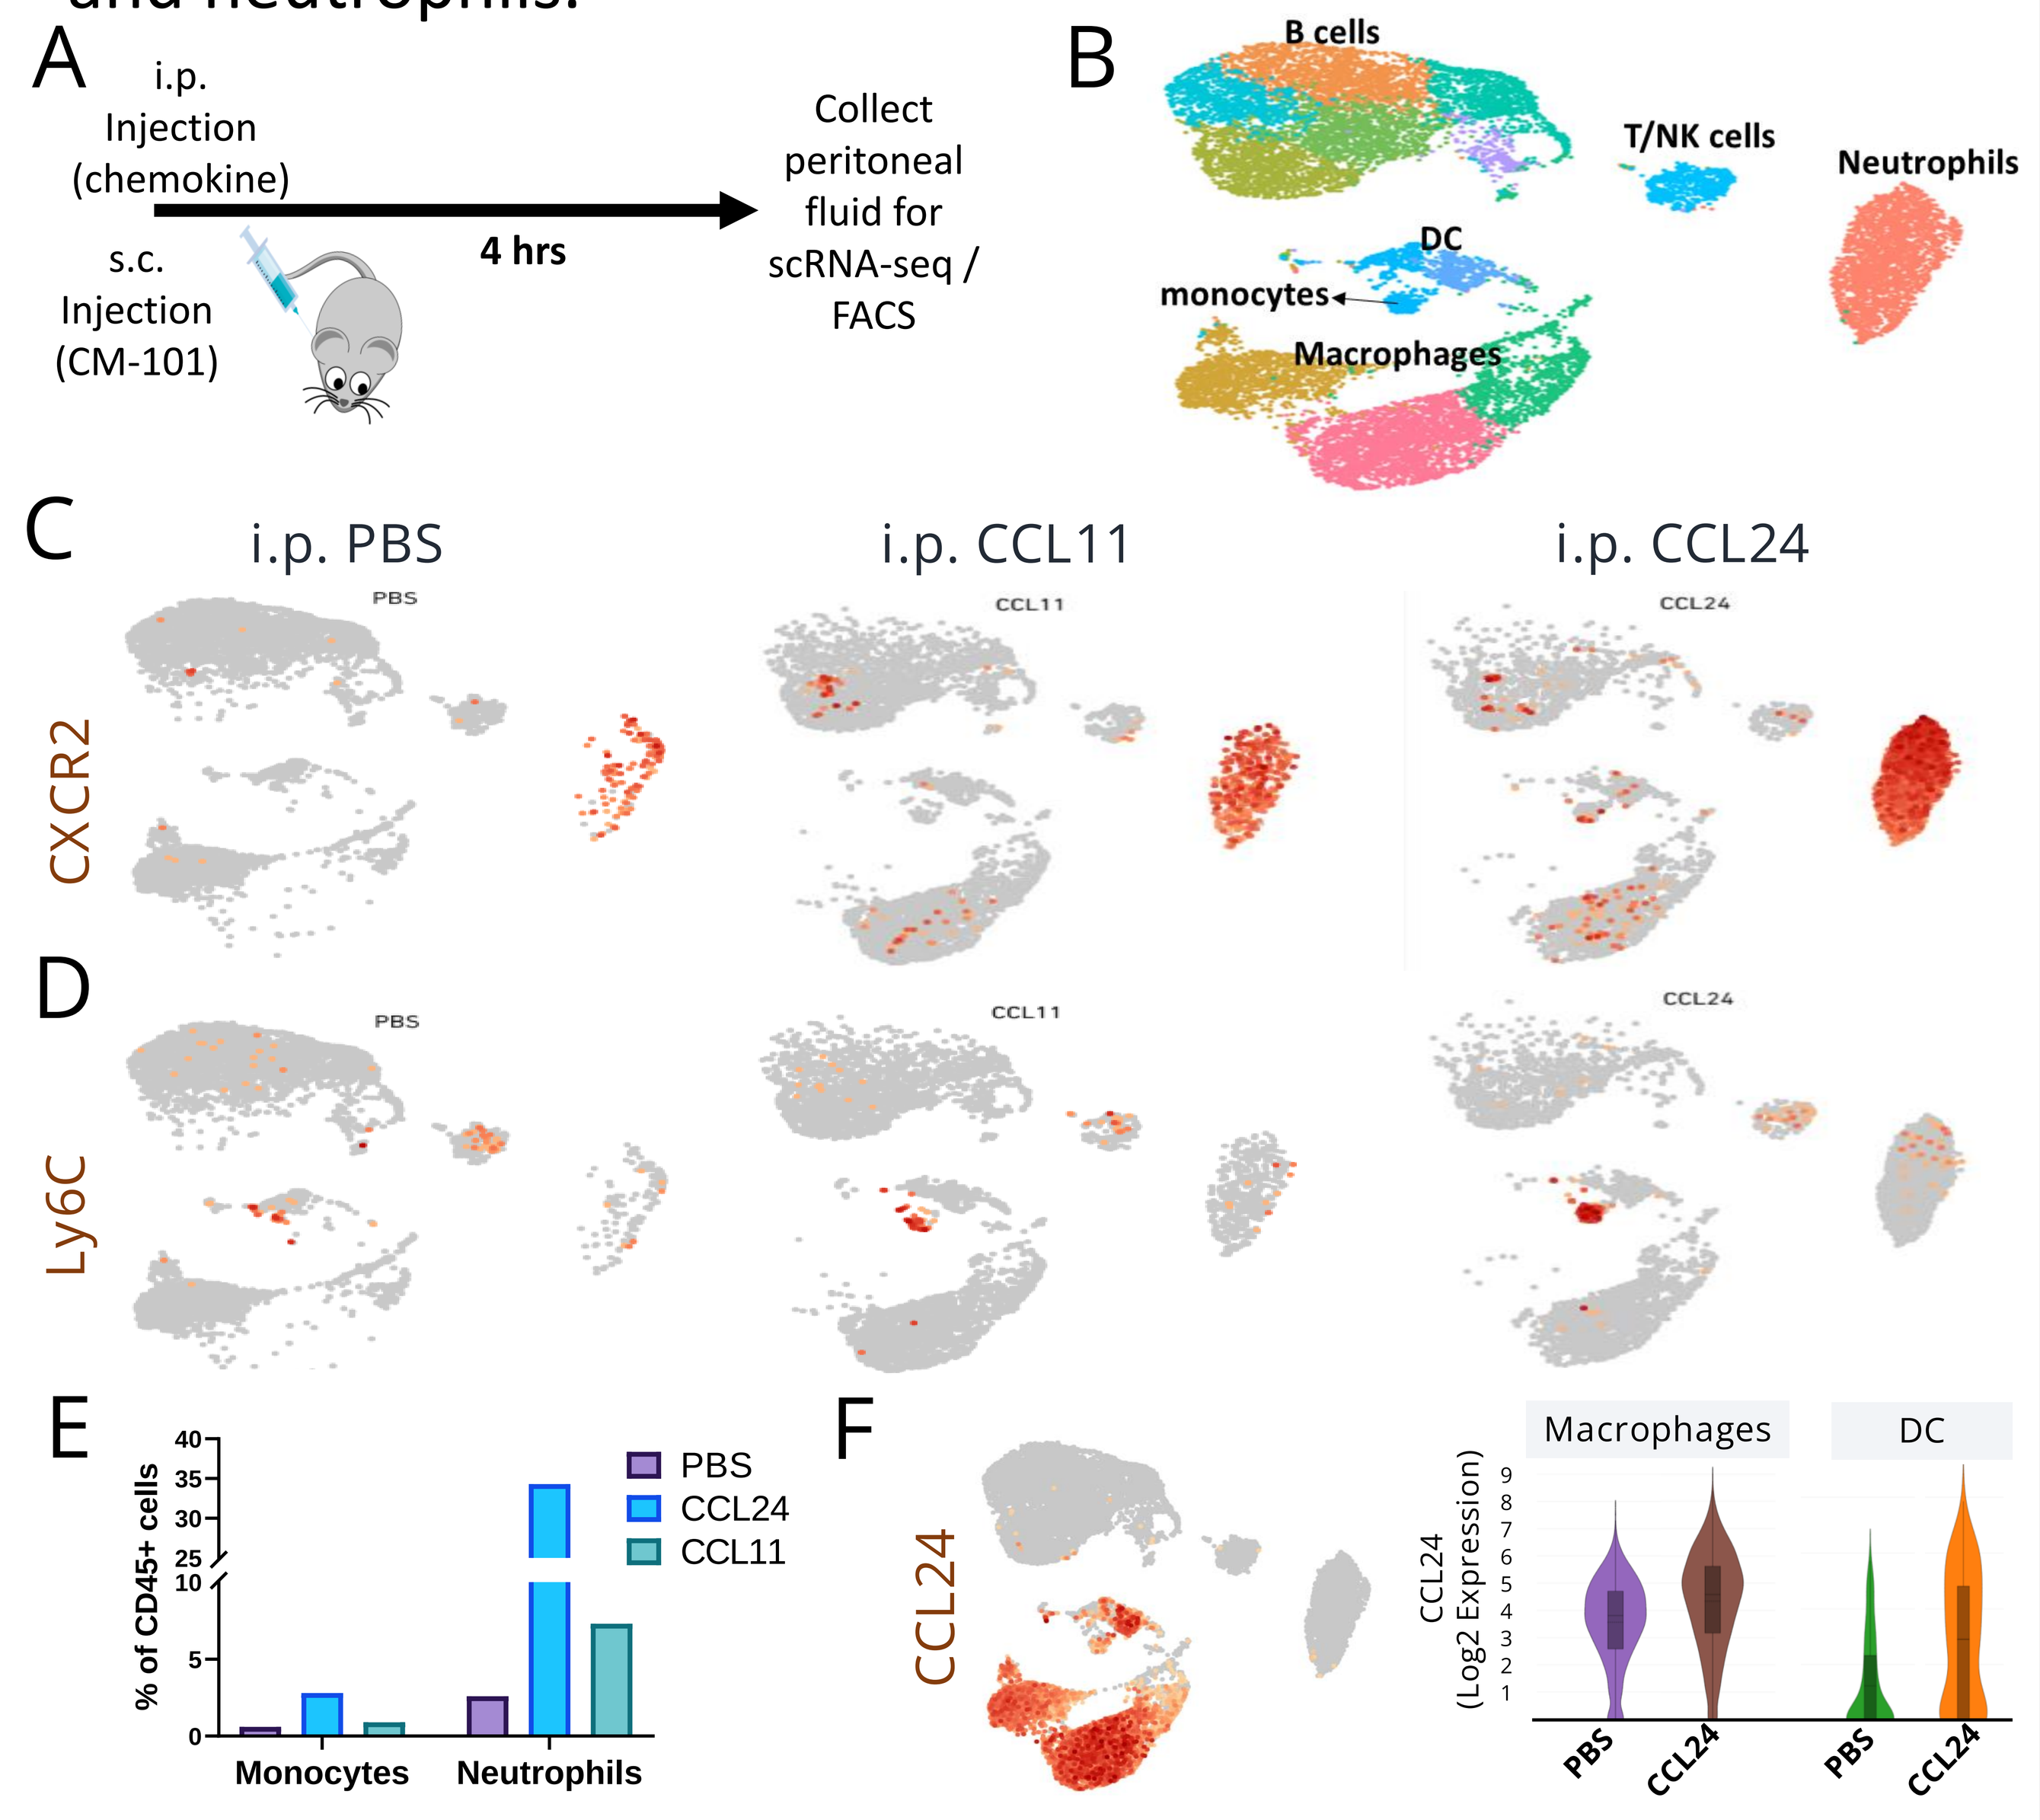

A
i.p. Injection (chemokine)
s.c. Injection (CM-101)
4 hrs
Collect peritoneal fluid for scRNA-seq / FACS
B
B cells
T/NK cells
Neutrophils
DC
monocytes
Macrophages
C
i.p. PBS
i.p. CCL11
i.p. CCL24
CXCR2
D
Ly6C
E
% of CD45+ cells
PBS
CCL24
CCL11
Monocytes
Neutrophils
F
CCL24
CCL24 (Log2 Expression)
Macrophages
DC
PBS
CCL24

scRNA-seq revealed increased recruitment of monocytes and neutrophils to the peritoneum after CCL24 injection. (A) Experiment scheme (B) UMAP representation with clusters identification. (C-D) Expression profiles of CXCR2, marker for Neutrophiles (C) and Ly6C1, marker for monocytes (D) following PBS, CCL11 or CCL24 i.p injections. (E) Quantification of number of monocytes and neutrophils. (F) Left panel, CCL24 expression profile. Right panel, quantification of CCL24 expression in macrophages and dendritic cells (DC).

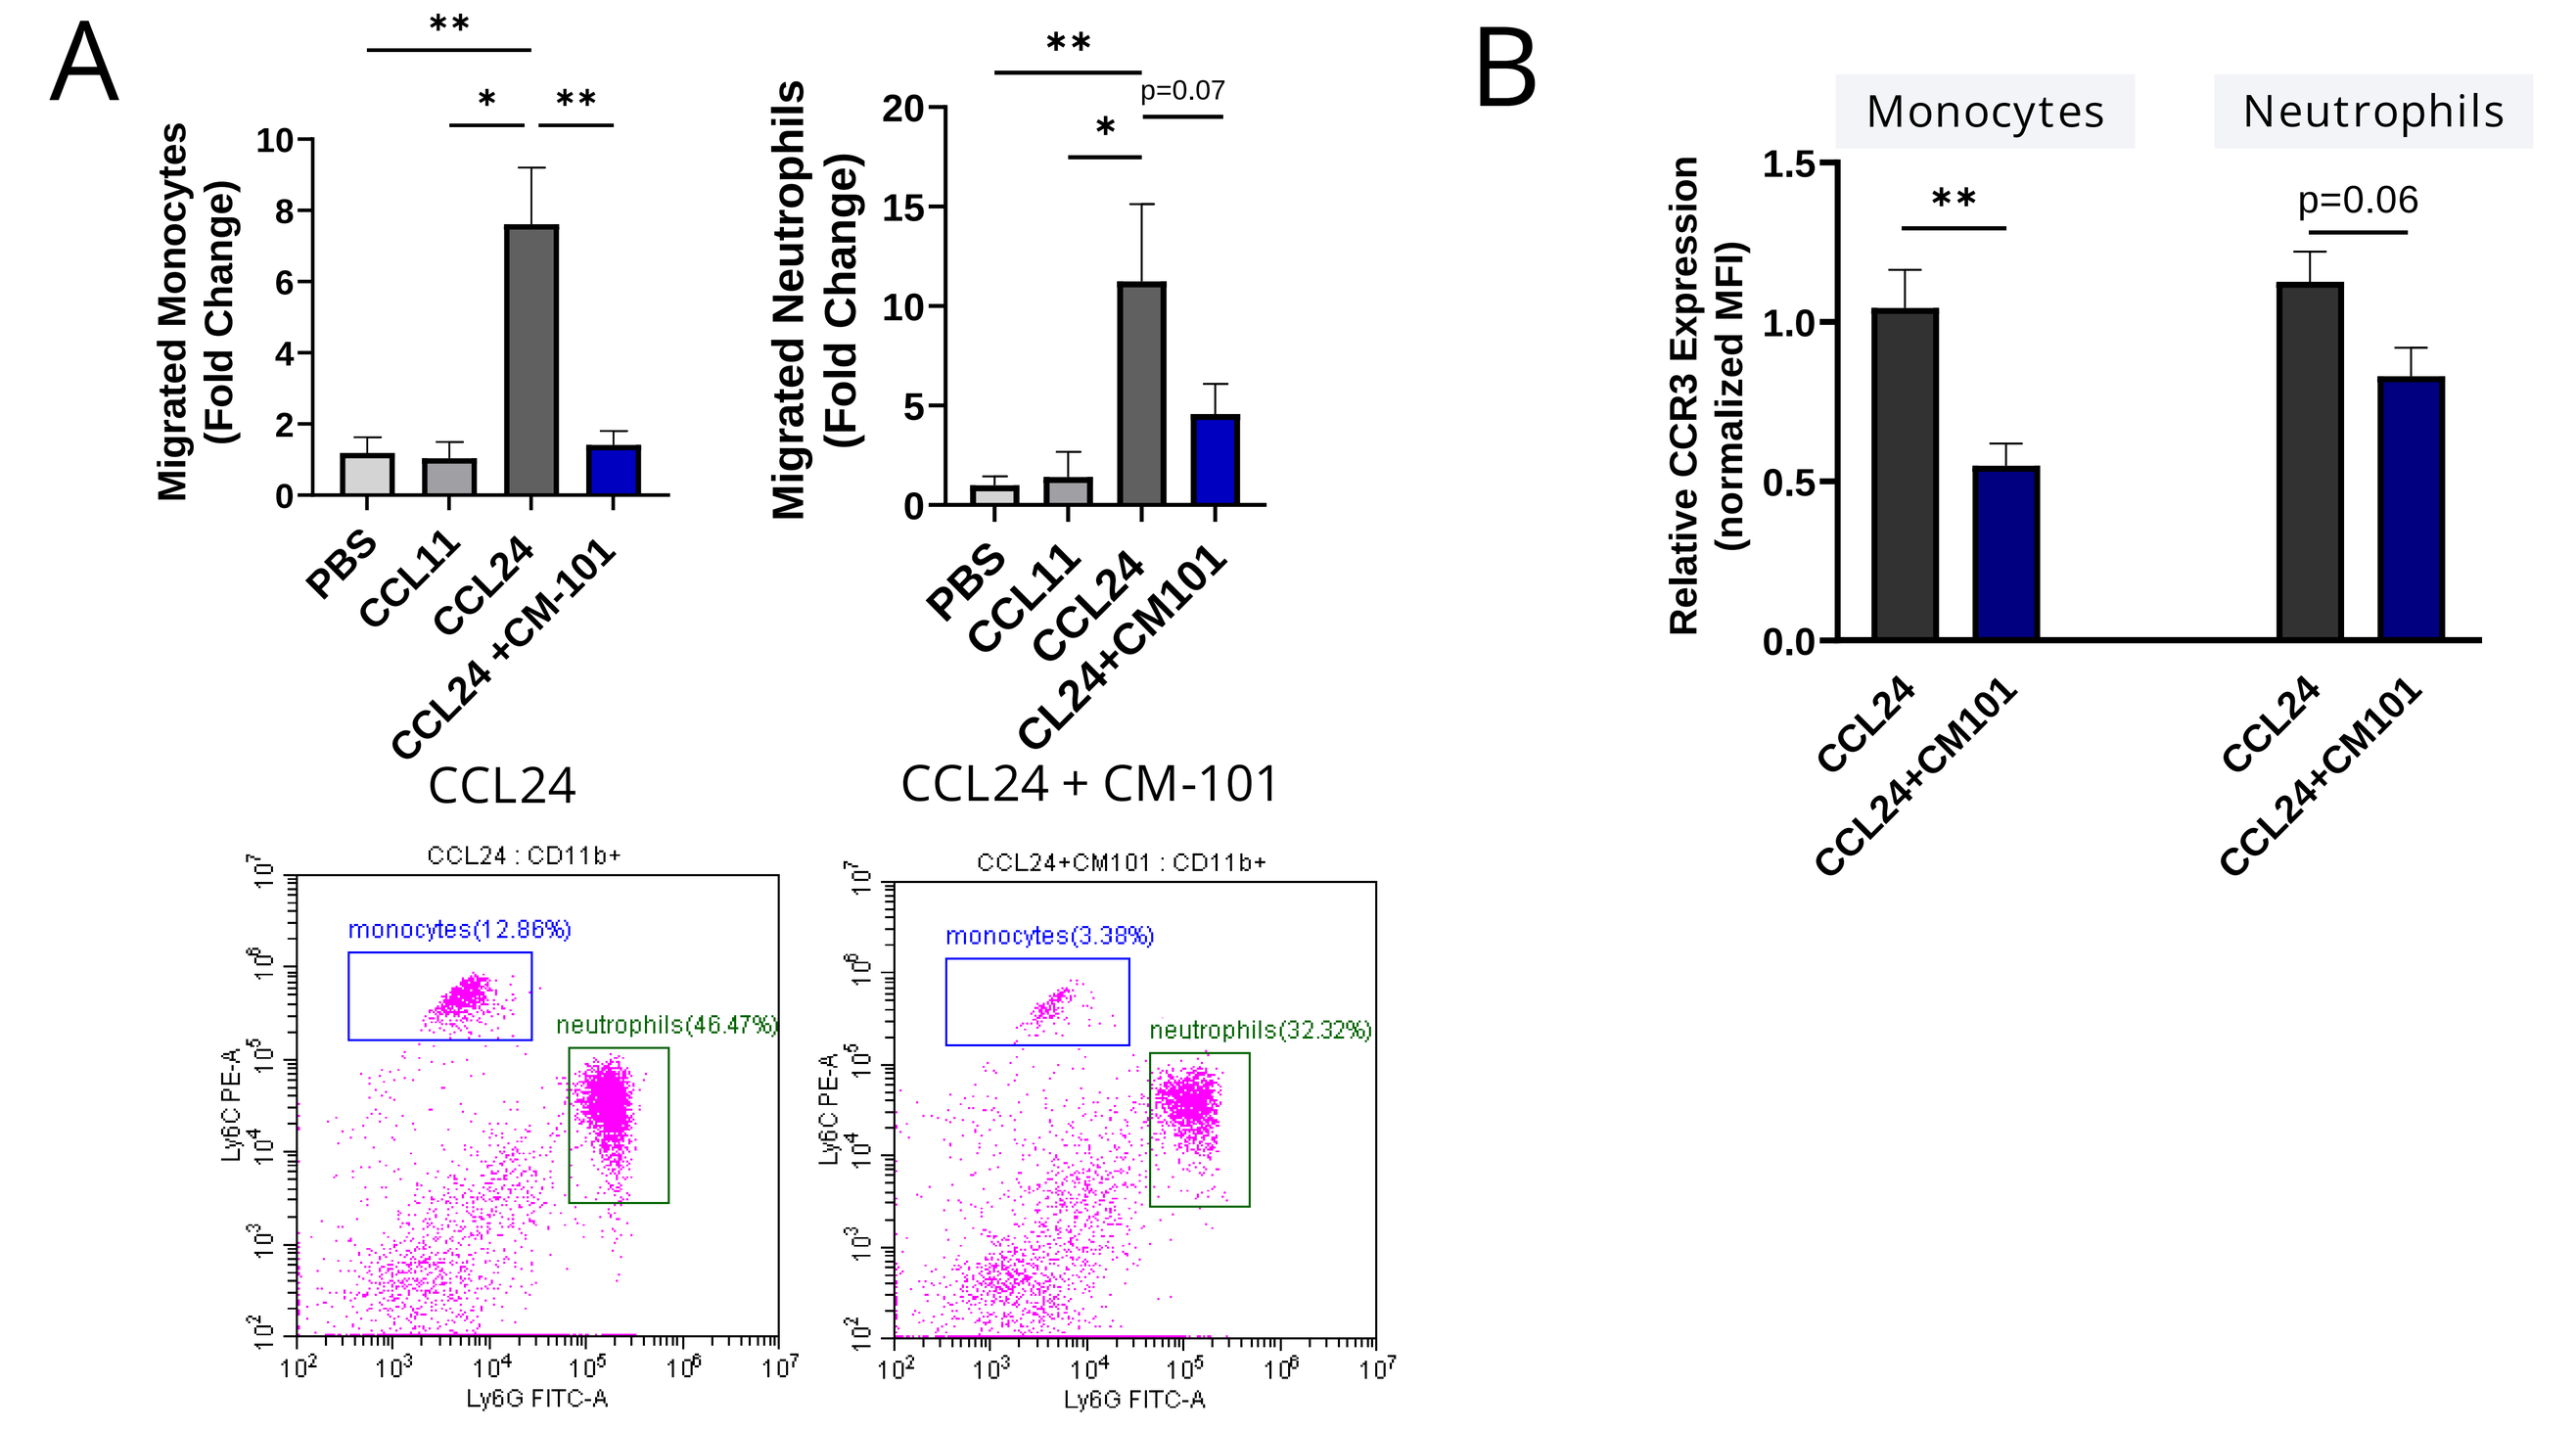

A
Migrated Monocytes (Fold Change)
Migrated Neutrophils (Fold Change)
p=0.07
PBS
CCL11
CCL24
CCL24 +CM-101
CCL24+CM101
CCL24
CCL24 + CM-101
monocytes(12.86%)
neutrophils(46.47%)
monocytes(3.38%)
neutrophils(32.32%)
Ly6G FITC-A
Ly6C PE-A
B
Monocytes
Neutrophils
Relative CCR3 Expression (normalized MFI)
p=0.06

CM-101 inhibits CCL24-dependent infiltration of immune cells to the peritoneum. (A) FACS analysis of monocytes and neutrophils. Upper panels, average quantification ± SEM. Lower panels, representative dot plots for CCL24 and CCL24+CM-101 treatments. (B) CCR3 expression (MFI) of neutrophils and monocytes is downregulated after CM-101 treatment.

"SPRING" PSC Clinical Trial

CM-101 Phase 2 Trial in Primary Sclerosing Cholangitis
Randomized, Double-Blind, Placebo-Controlled

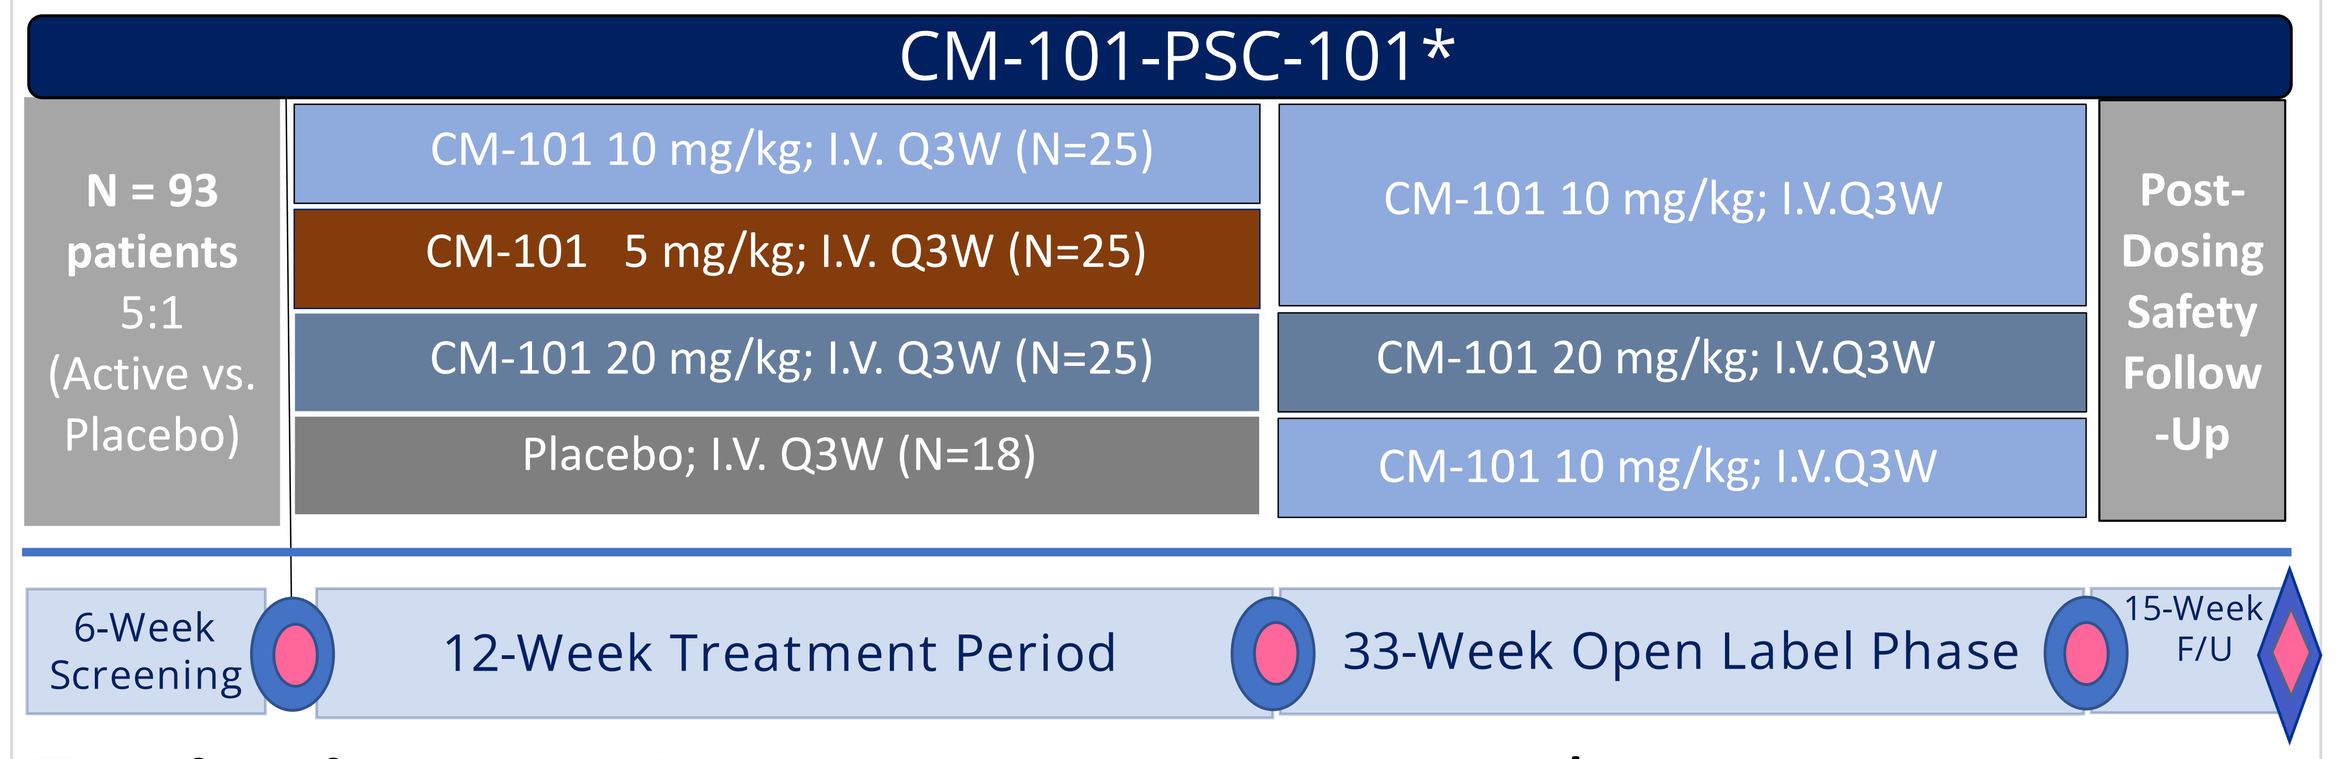

CM-101-PSC-101*
N = 93 patients 5:1 (Active vs. Placebo)
CM-101 10 mg/kg; I.V. Q3W (N=25)
CM-101 5 mg/kg; I.V. Q3W (N=25)
CM-101 20 mg/kg; I.V. Q3W (N=25)
Placebo; I.V. Q3W (N=18)
CM-101 10 mg/kg; I.V.Q3W
CM-101 20 mg/kg; I.V.Q3W
CM-101 10 mg/kg; I.V.Q3W
Post-Dosing Safety Follow-Up
6-Week Screening
12-Week Treatment Period
33-Week Open Label Phase
15-Week F/U

Territories: US, UK, Germany, Spain, Israel

Key Enrollment Criteria:
- PSC patients with large duct disease of >24 weeks duration
- ALP > 1.5 ULN
- Stable IBD allowed
- Stable UDCA treatment allowed

Outcome Measures:

Primary - Safety

Secondary - Change from baseline to Week15 in:
- Serum alkaline phosphatase
- ELF score
- Fibrotic biomarkers/liver enzymes-AST, ALT, Pro-C3, Pro-C5&Fibroscan
- Pharmacokinetics
- Anti-Drug Antibody
- Pharmacodynamic parameters

ClinicalTrials.gov Identifier: NCT04595825

Conclusions

- Monocytes and neutrophils are major players in PSC pathophysiology.
- Two in-vivo models for immune cell trafficking demonstrate clear CCL24-dependent recruitment of these immune cells. These inflammatory effects are unique to CCL24 compared with CCL11.
- CM-101, a first-in-class CCL24 neutralizing mAb, demonstrated an anti-inflammatory effect by interfering with migration of these cells to the damaged biliary area in a PSC model.
- CCL24 is a promising therapeutic target for PSC treatment. CM-101 is currently being tested in a Phase 2 study in PSC patients.

Disclosures

RG, IV, MSS, AK, OL and AM are employees of Chemomab
AP is a consultant to Chemomab

CONTACT INFORMATION: ilan.v@chemomab.com