Patient Proteomic Data and Mouse Model Reinforce the Proinflammatory Role of CCL24 in Cholestatic Disease

Raanan Greenman[1], Tom Snir[1], Revital Aricha[1], Douglas Thorburn[2], Massimo Pinzani[3], John Lawler[1], Ilan Vaknin[1], Matthew Frankel[1]

[1]Chemomab Therapeutics, Israel & United States

[2]Royal Free London NHS Foundation Trust, United Kingdom

[3]UCL Institute of Immunity & Transplantation, United Kingdom

INTRODUCTION

Primary Sclerosing Cholangitis (PSC) is a chronic liver disease that is marked by the presence of a damaged peribiliary space. In this space, a complex interplay occurs between immune cells, reactive biliary epithelial cells and fibroblasts, resulting in elevated levels of cytokines, chemokines, and other secreted factors that contribute to the inflammatory process. Specifically, the chemokine CCL24 has been implicated in driving these self-reinforcing mechanisms in fibrotic-inflammatory diseases. Preclinical studies have demonstrated that blocking CCL24 can attenuate the progression of PSC in animal models. Specifically, monocytes and neutrophils are major players in PSC pathophysiology, and studies using in in-vivo models for immune cell trafficking have shown that the chemokine CCL24 plays a role in recruiting these immune cells.

AIM

We aim to study the role of CCL24, a chemokine that promotes immune cell trafficking and activation as well as pro-fibrotic activities at inflammatory peribiliary areas in PSC pathophysiology.

- We examined the effect of CCL24 blockade using CM-101, a CCL24 neutralizing monoclonal antibody, on the trafficking of immune cells to the diseased biliary area in ANIT induced cholestasis model.
- We explored the association of CCL24 with biological pathways that are related to immune-cell chemotaxis in patients' serum.

METHOD

- Chronic α-naphthylisothiocyanate (ANIT)-induced cholestasis mouse model was employed. Mice were fed with an ANIT diet (0.05%) for 4 weeks and treated twice a week with either 5 mg/kg CM-101 or a vehicle control during weeks 2 through 4. Liver inflammation was evaluated by immunohistochemistry staining for various immune markers. Liver fibrosis and biliary hyperplasia were evaluated by hematoxylin & eosin (H&E) and Pan-CK staining, respectively.
- For human serum proteomics, sera from healthy controls (n = 30) and patients with PSC (n = 45) were evaluated for expression of immune-migration related proteins using the Olink proximity extension assay. Demographics and enhanced liver fibrosis (ELF) scores were captured. Average expression of proteins related to pathways associated with immune cell migration was compared between: (1) healthy controls and patients with PSC, (2) high and low serum levels of CCL24 (stratified by the mean) within patients with PSC, and (3) high and low ELF score (stratified by a score of 9.8) within patients with PSC.

RESULTS

ANIT-induced cholestasis animal model

CCL24 blockade with CM-101 mAb reduces liver fibrosis and biliary hyperplasia, and neutrophil and macrophage accumulation in the peribiliary area

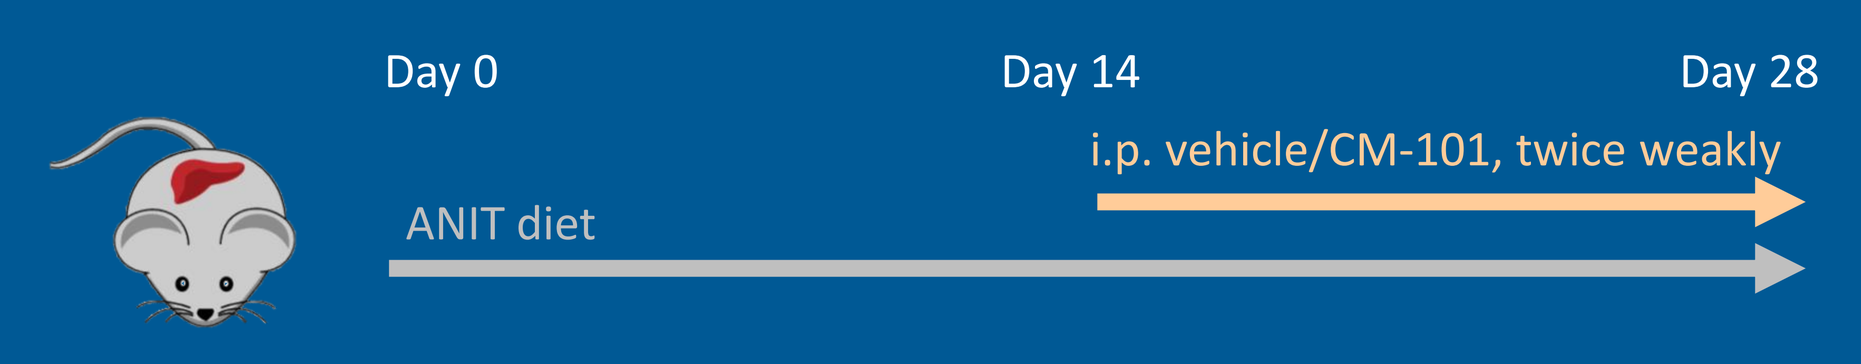
Day 0
Day 14
Day 28
i.p. vehicle/CM-101, twice weakly
ANIT diet

CM-101 treatment reduces neutrophil and macrophage accumulation in the peribiliary area

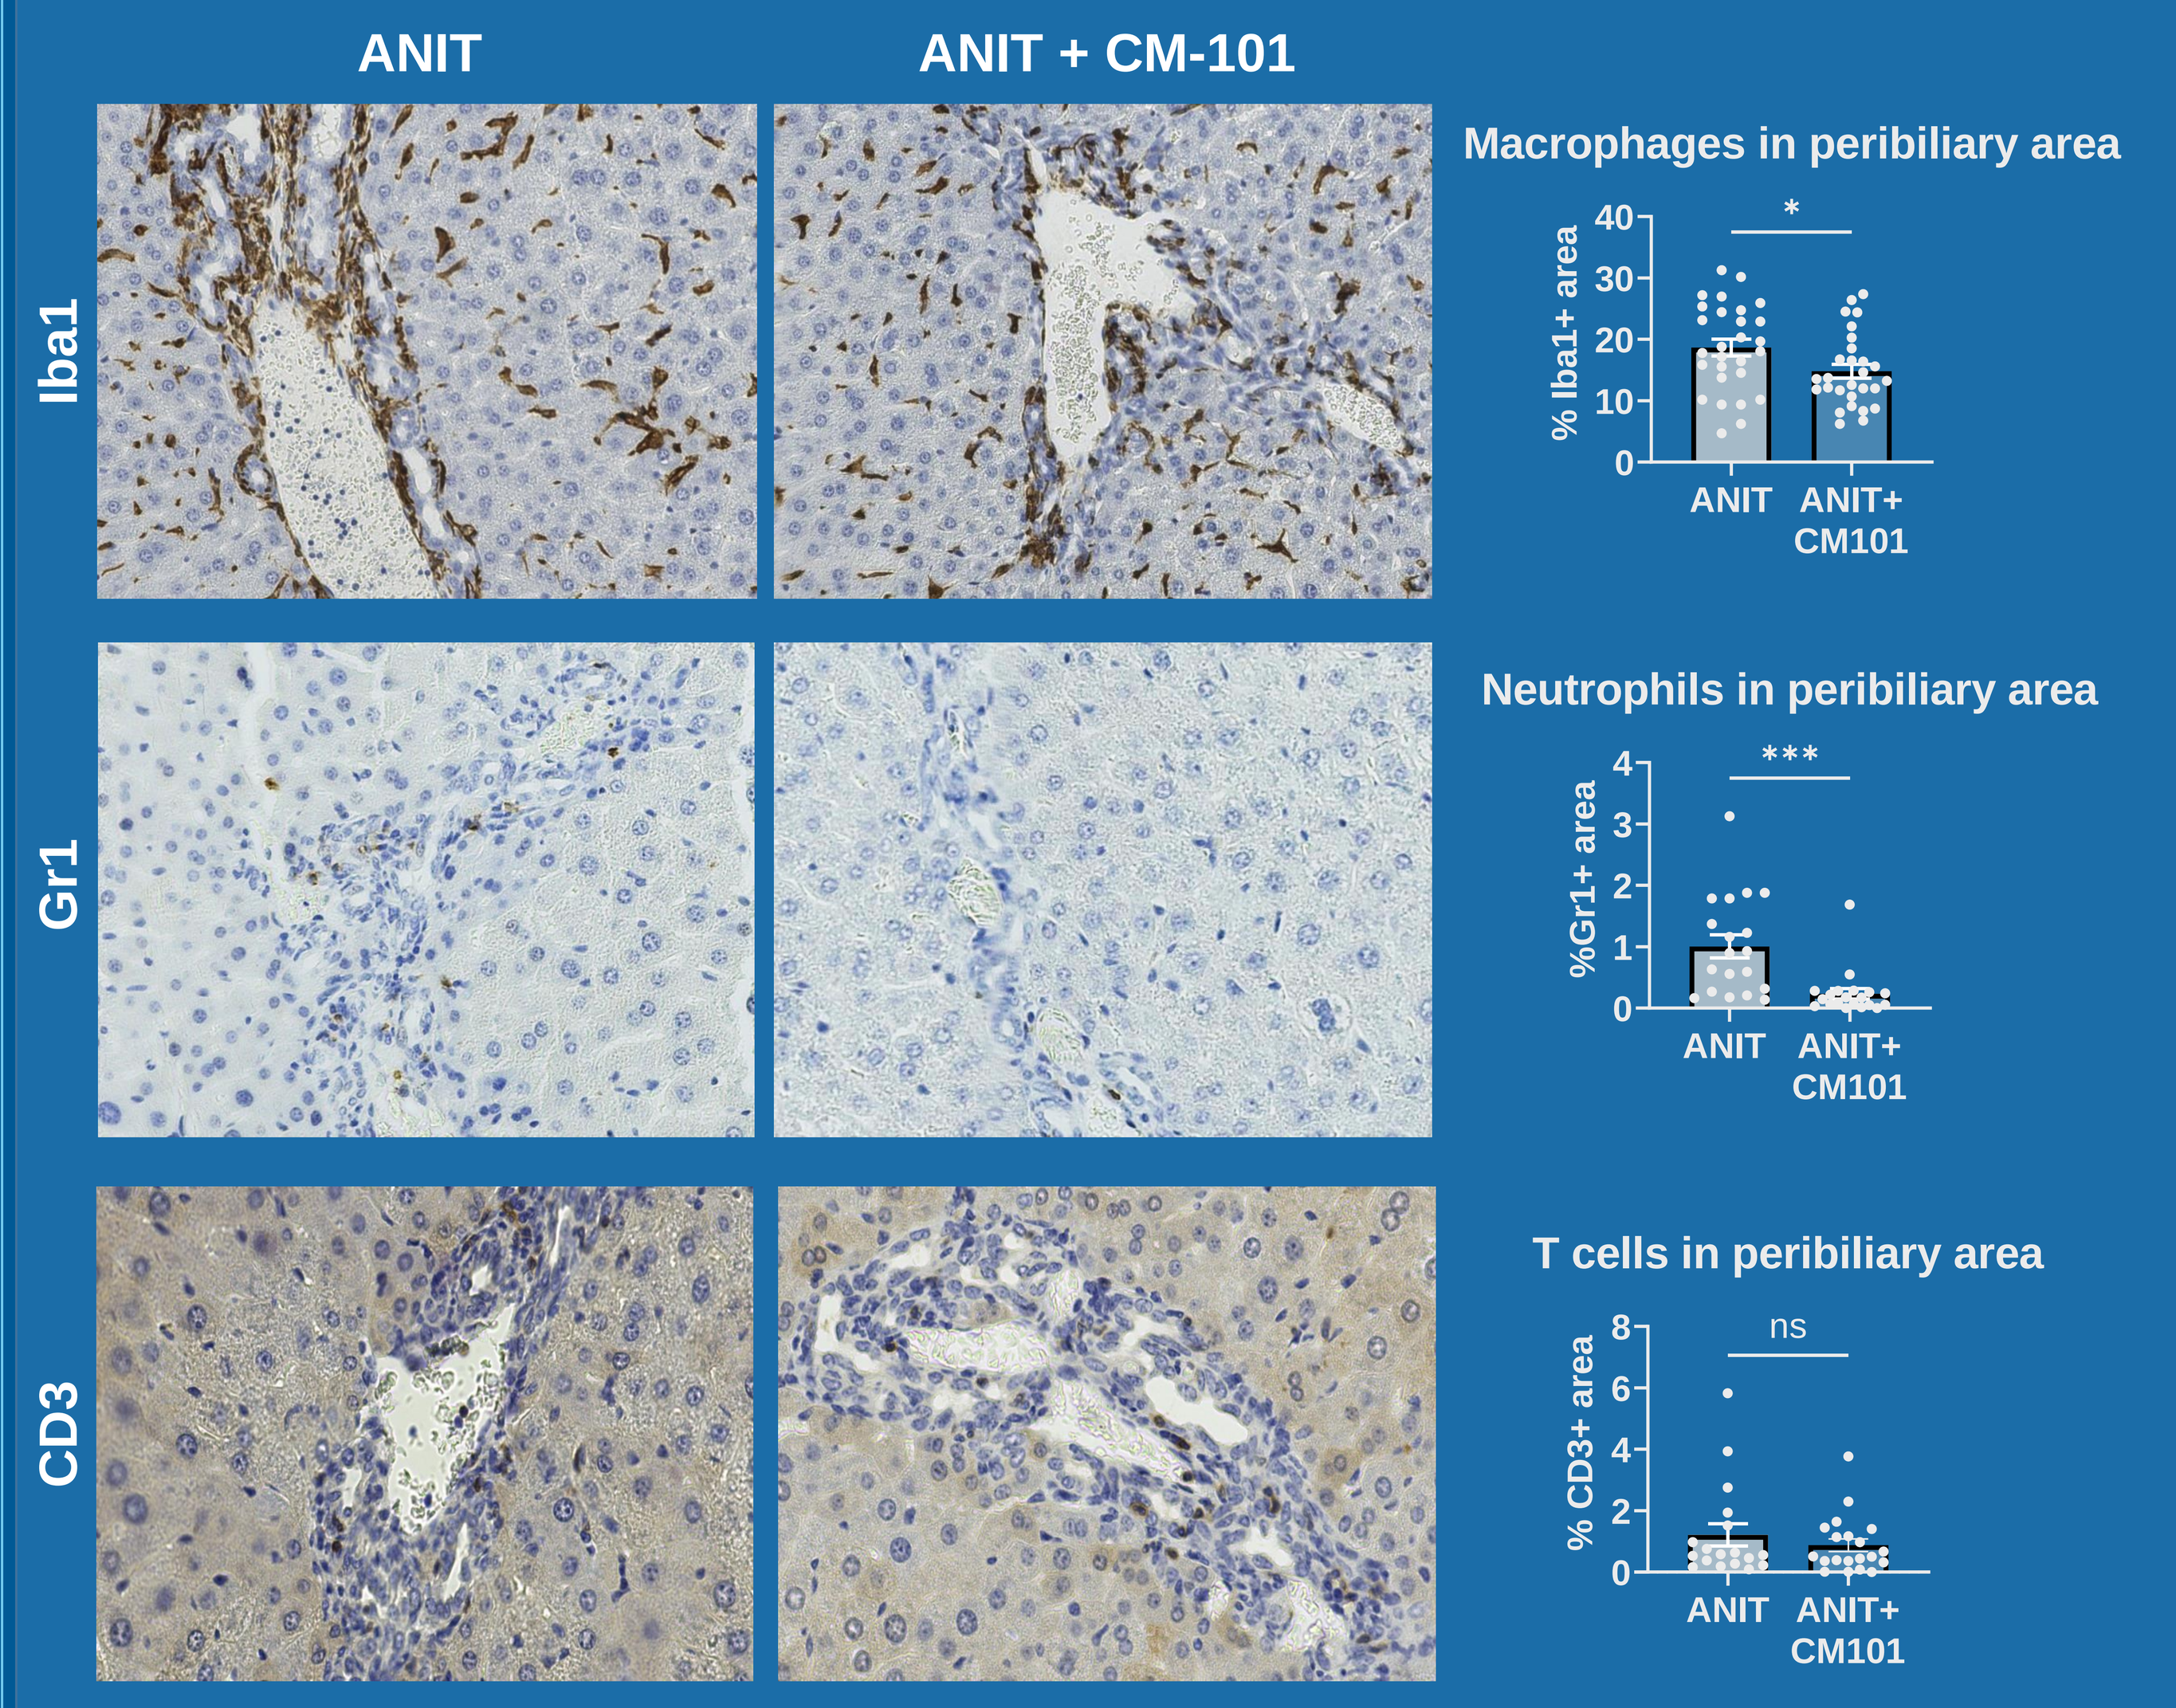
ANIT
ANIT + CM-101
Iba1
Gr1
CD3
Macrophages in peribiliary area
% Iba1+ area
Neutrophils in peribiliary area
%Gr1+ area
T cells in peribiliary area
% CD3+ area
ns
ANIT
ANIT+ CM101

Liver staining for macrophage (Iba1), neutrophil (Gr1) and T cell (CD3) accumulation in the peribiliary area demonstrates specific recruitment of neutrophils and monocytes, whereas T cells remain unchanged. Left panels, representative images; right panels, quantification - mean ± SEM

CM-101 treatment reduces liver fibrosis and biliary hyperplasia

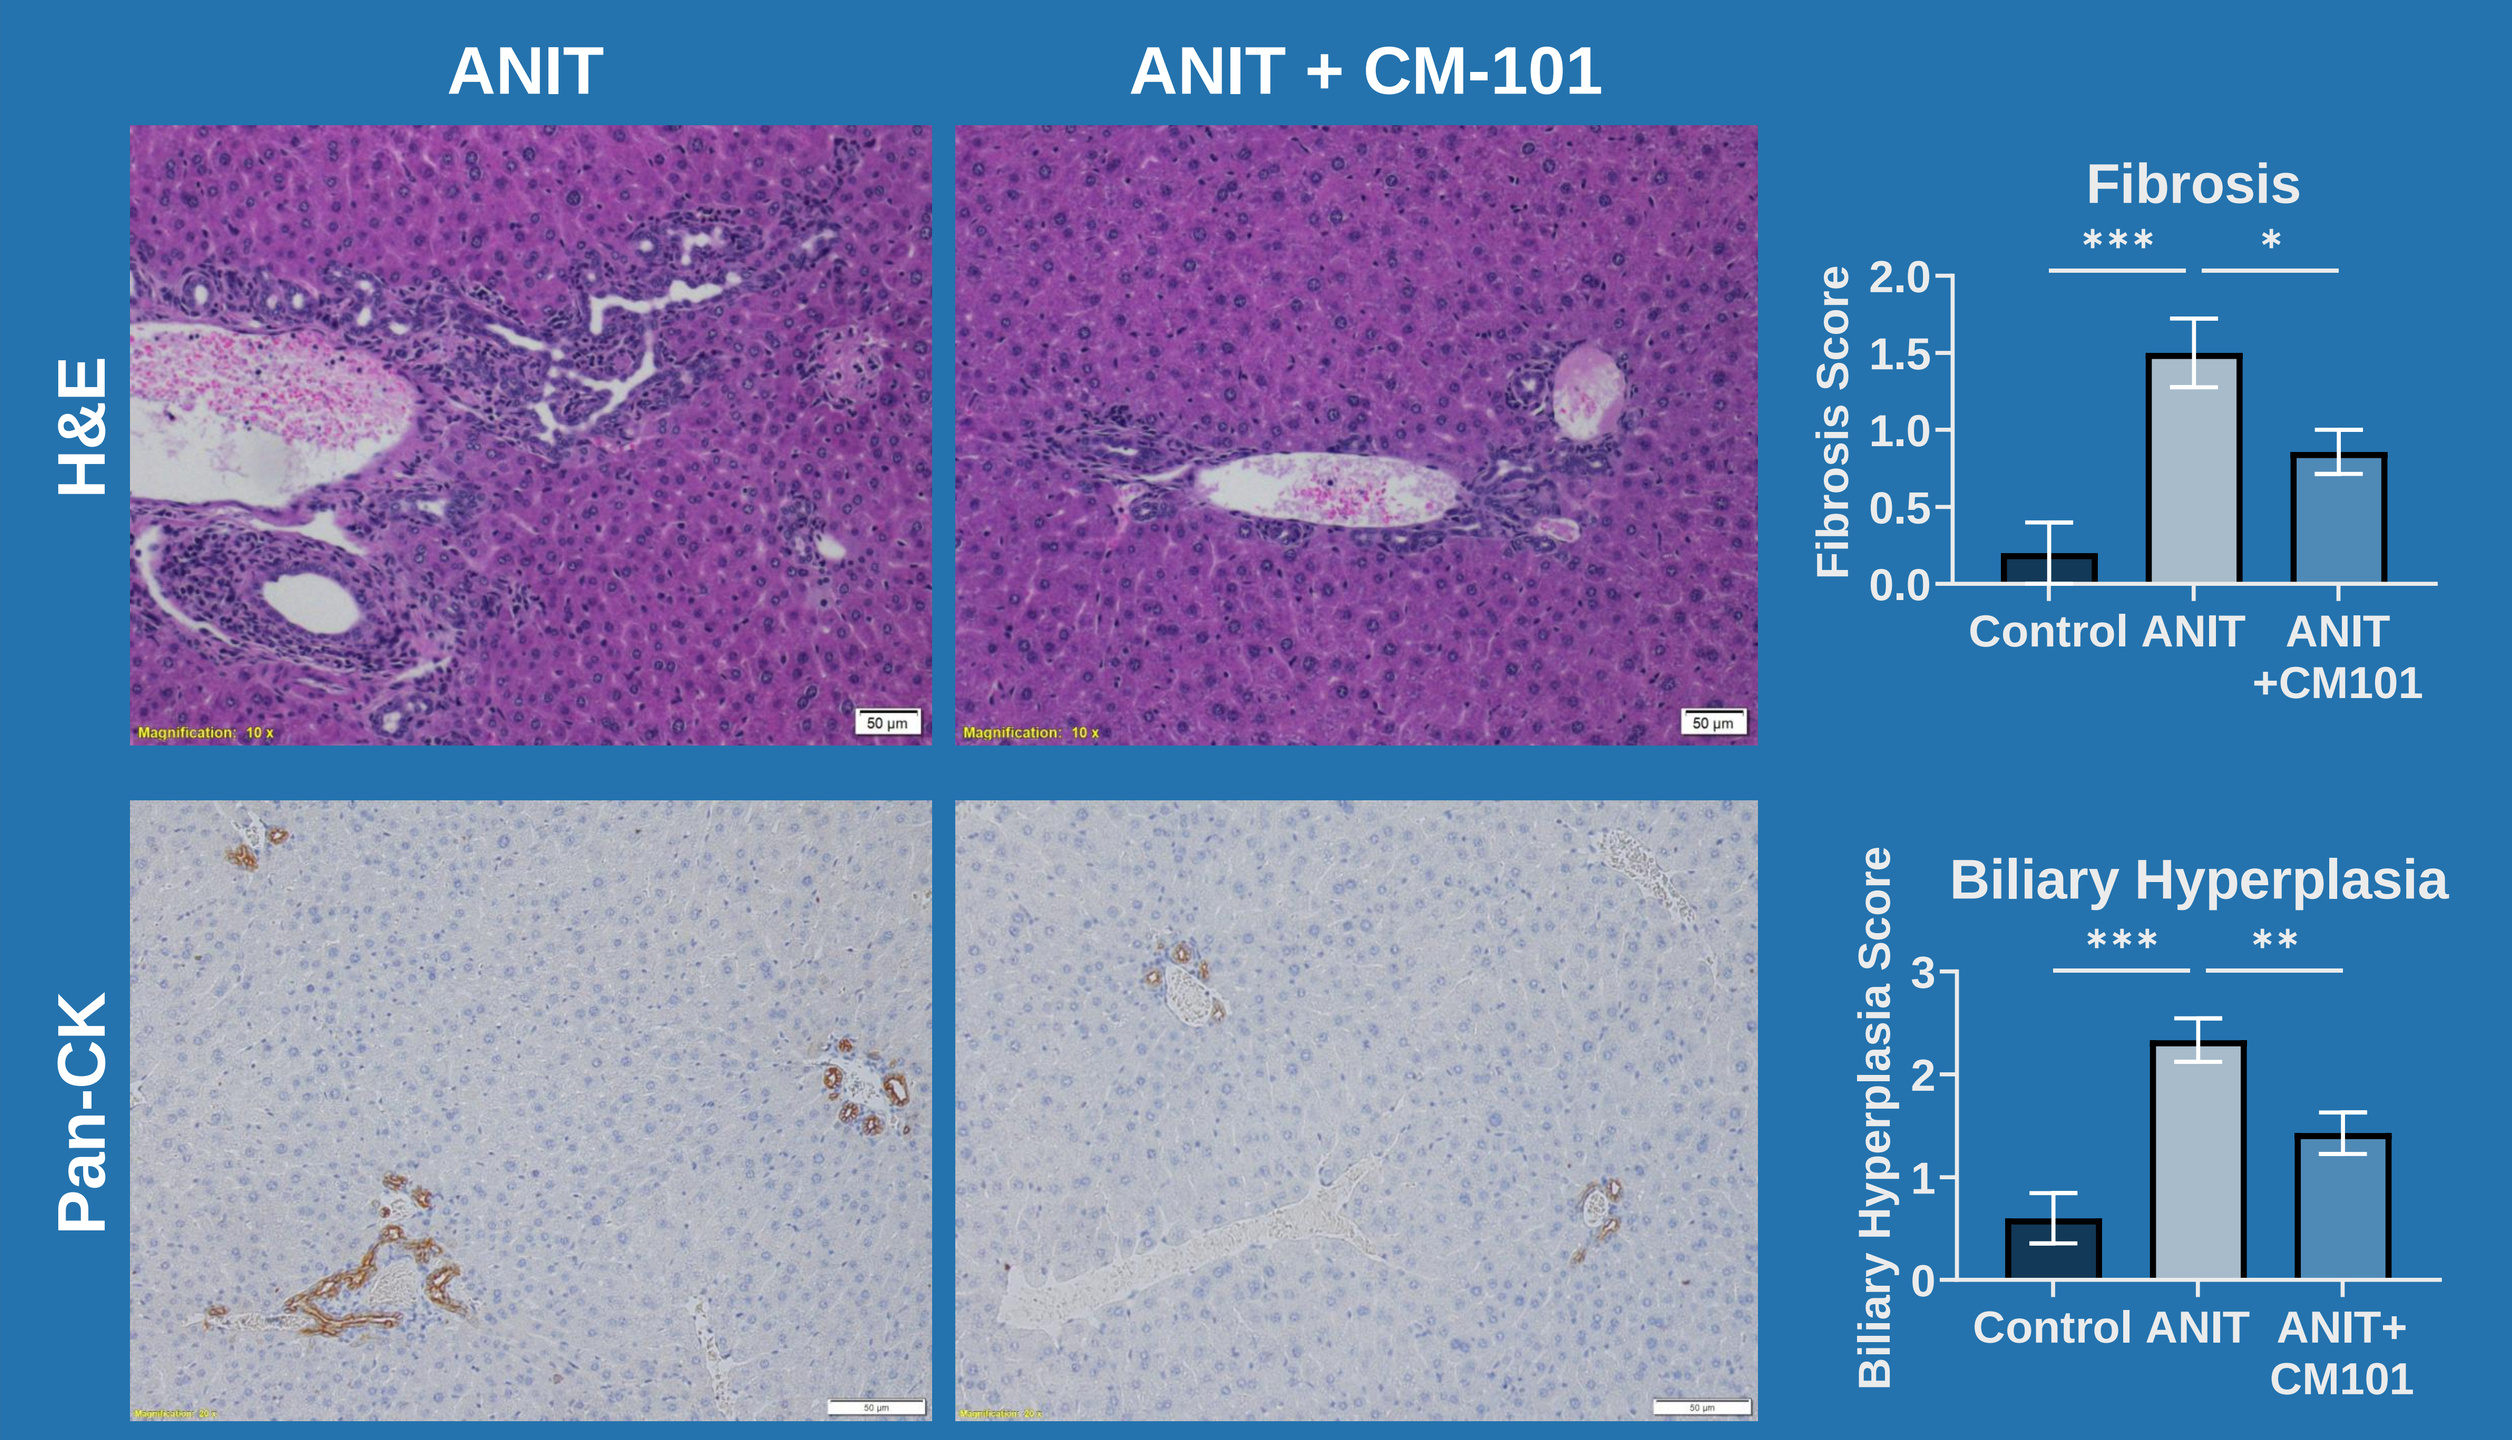
ANIT
ANIT + CM-101
H&E
Pan-CK
Fibrosis
Fibrosis Score
Biliary Hyperplasia
Biliary Hyperplasia Score
Control
ANIT
ANIT +CM101

Liver staining for fibrosis (H&E) and cholangiocyte (Pan-CK). Left panels, representative images; right panels, quantification ± SEM.

Neutrophil and macrophage peribiliary accumulation correlates with liver fibrosis and biliary hyperplasia

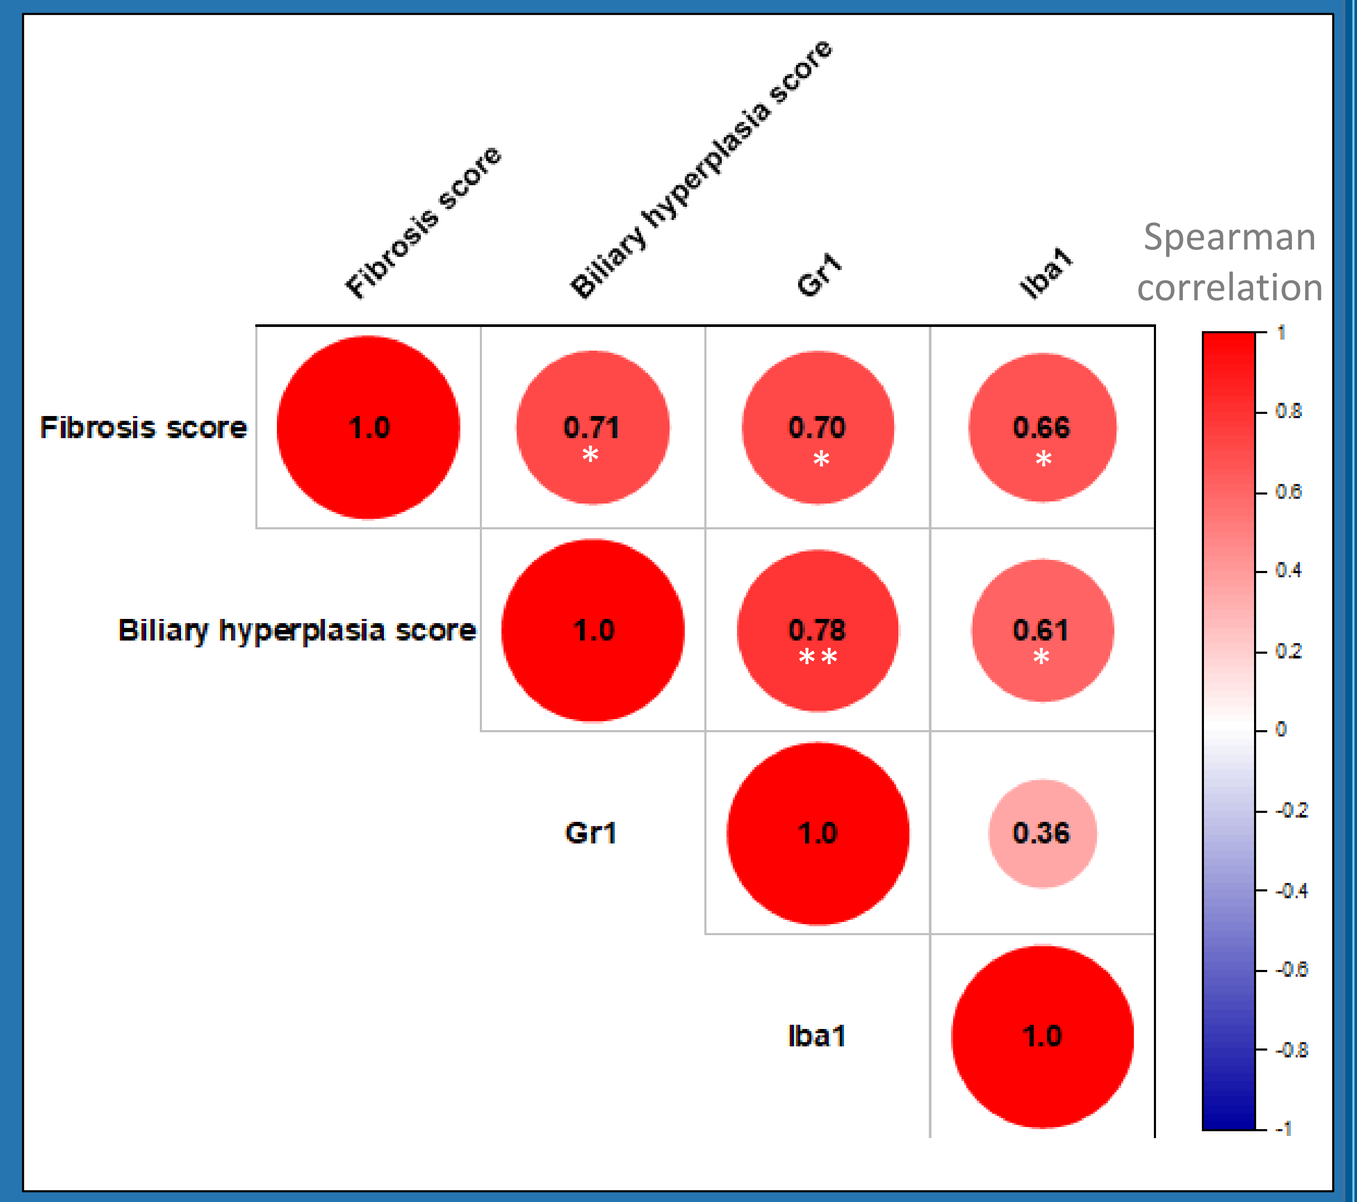
Fibrosis score
Biliary hyperplasia score
Gr1
Iba1
Spearman correlation
1.0
0.71
0.70
0.66
0.78
0.61
0.36

Correlation matrix between fibrosis score, biliary hyperplasia score, Iba1 positive stain area and Gr1 positive stain area. Correlation is performed on CM101 treated and untreated mice.

PSC patients: Serum proteomics

High CCL24 serum levels associate with upregulation of monocyte and neutrophil chemotaxis pathways in PSC

	N	Age [y], median (range)	Duration since diagnosis [y], median (range)	Male, n (%)	IBD any, n (%)	ALP [U/L], median (range)	ALT [U/L], median (range)	AST [U/L], median (range)	Bilirubin [mg/dL], median (range)	Fibroscan, n, median (range)	ELF, n, median (range)
Healthy controls	30	23.5 (18-38)	NA	30 (100)	0 (0)	74 (42-106)	16.5 (7-45)	19 (12-34)	11 (7-18)	NA	NA
PSC	45	45 (18-76)	4.9 (0-25.3)	26 (58)	30 (67)	246 (52-1064)	70 (10-796)	54 (15-919)	12 (3-41)	43, 10.1 (5.0-17.3)	33, 9.95 (7.85-12.84)

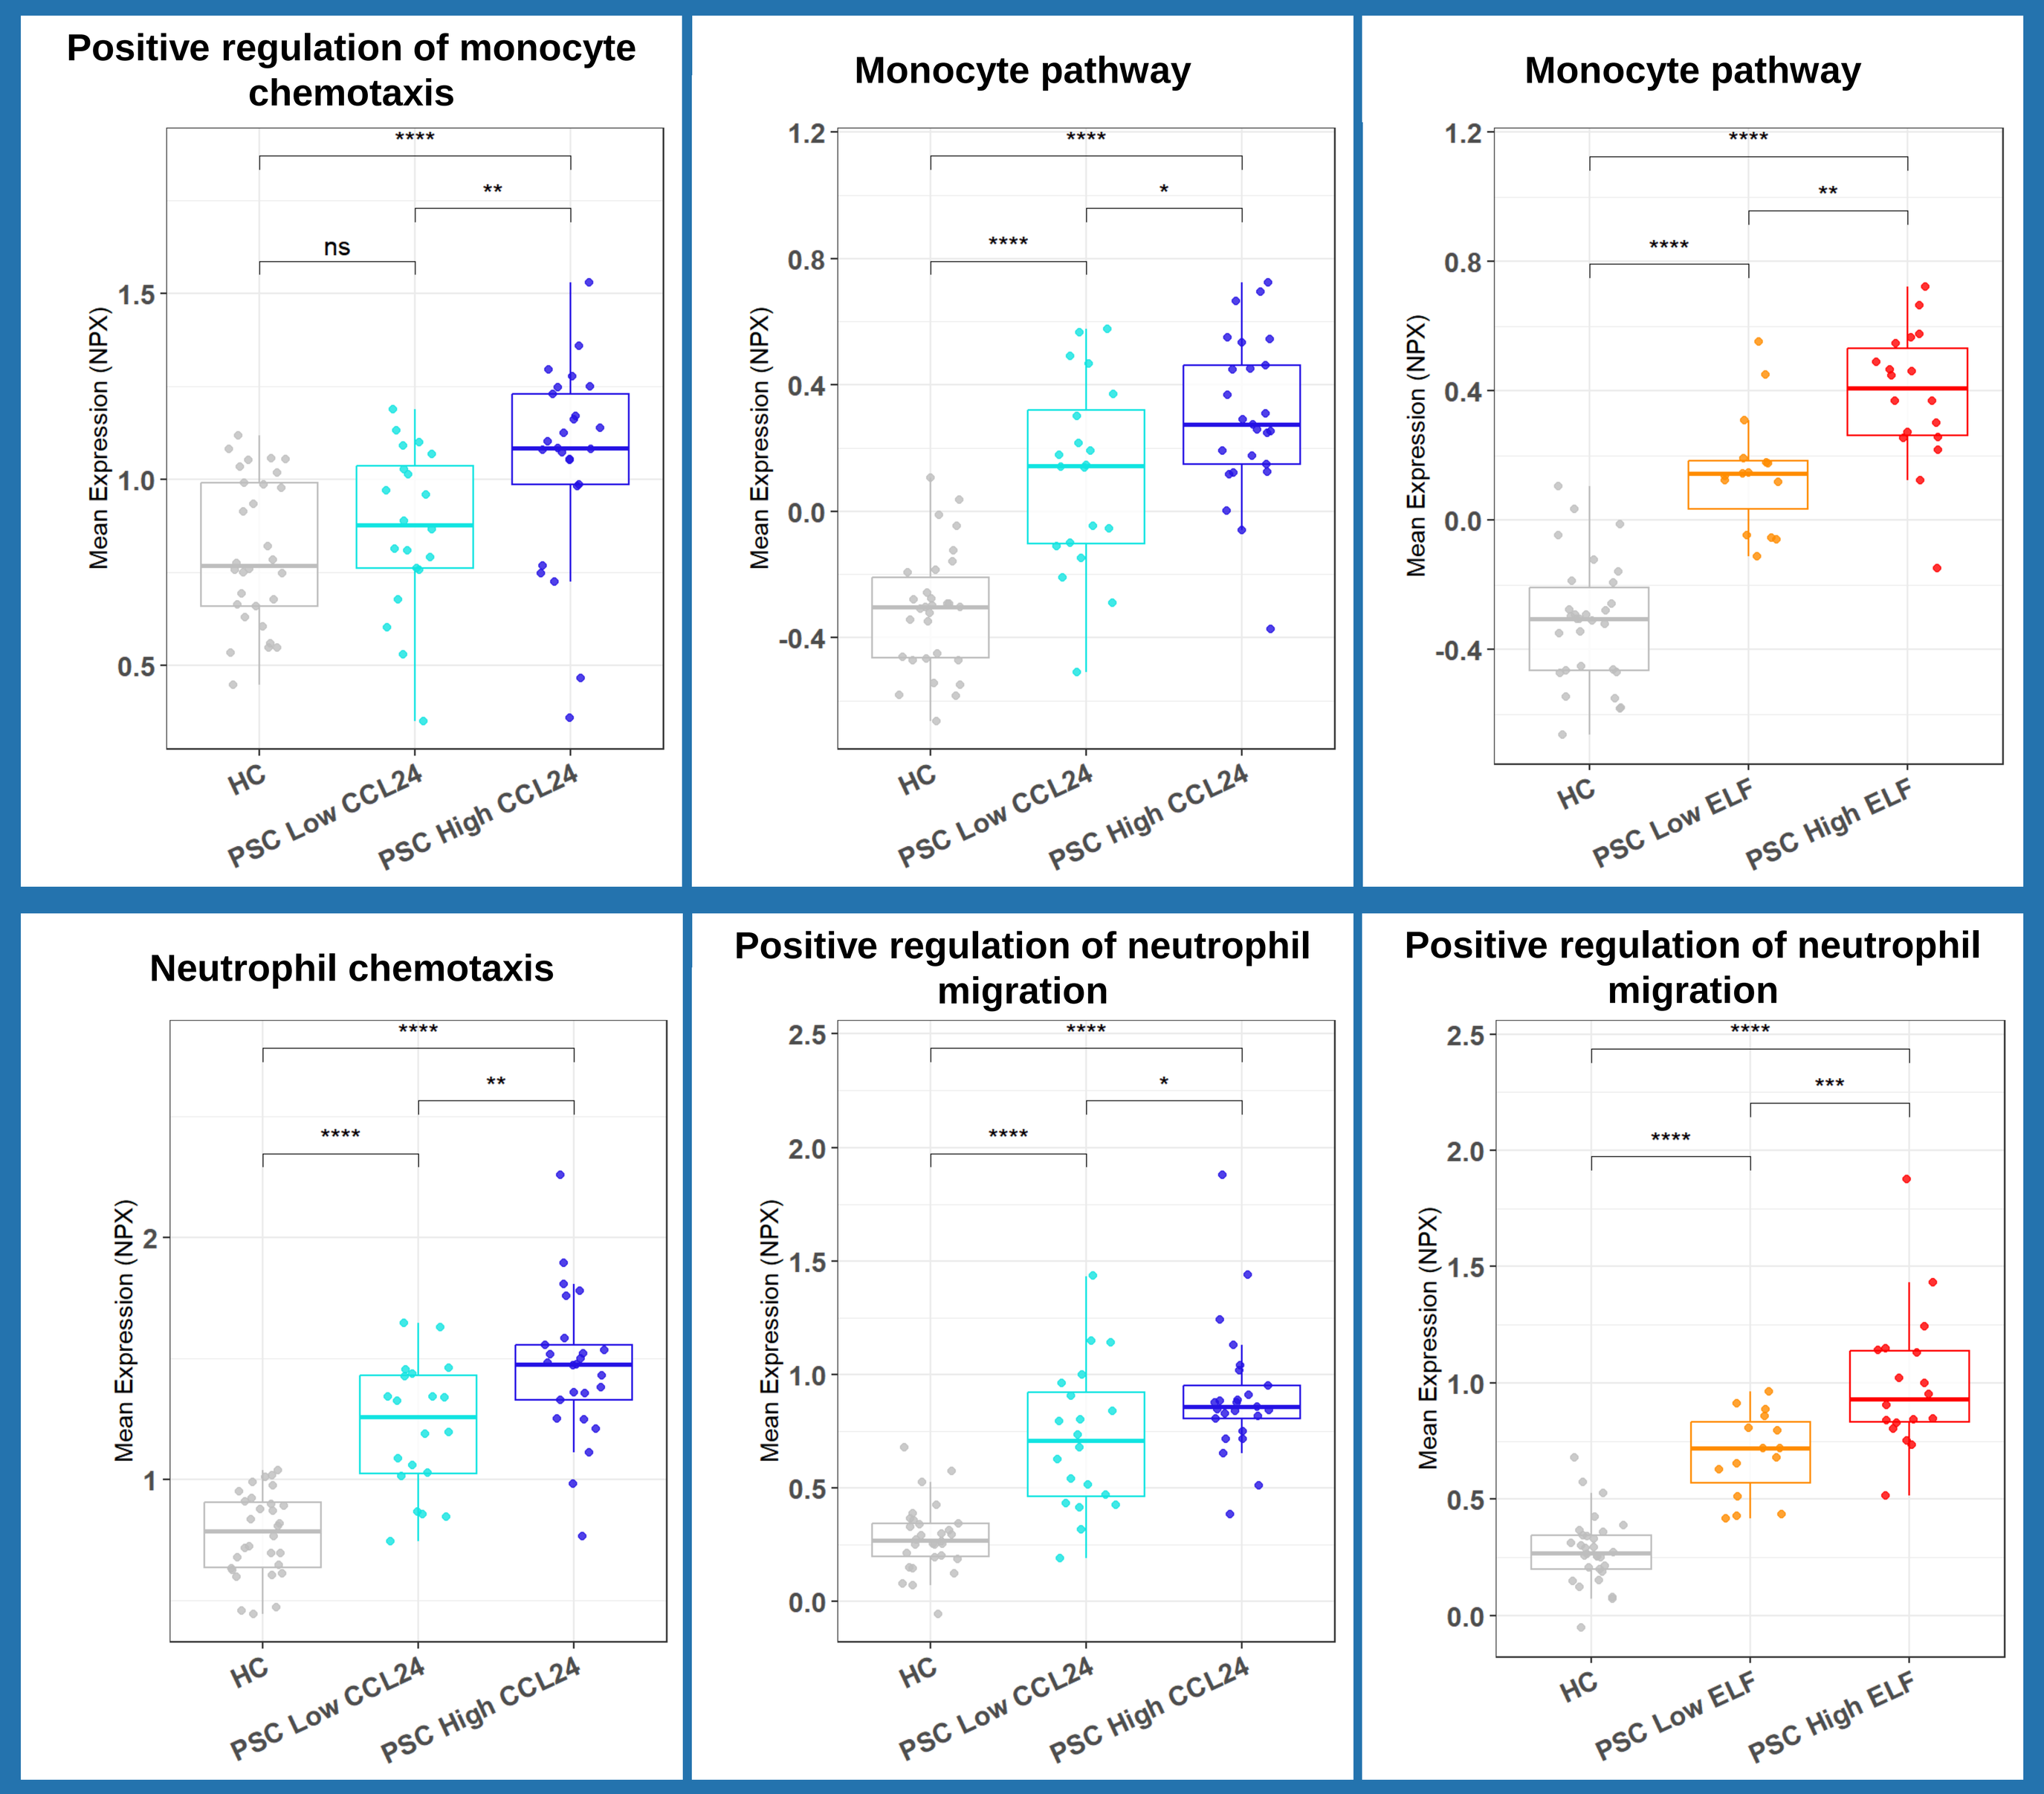
Positive regulation of monocyte chemotaxis
Monocyte pathway
Monocyte pathway
Neutrophil chemotaxis
Positive regulation of neutrophil migration
Positive regulation of neutrophil migration
Mean Expression (NPX)
HC
PSC Low CCL24
PSC High CCL24
PSC Low ELF
PSC High ELF
ns

Expression of pathway-specific plasma protein signatures in healthy controls versus PSC patients with low and high CCL24 serum levels (stratified by the mean), or versus PSC patients with moderate and severe disease (cutoff of 9.8 ELF score). Boxes represent interquartile ranges with medians. HC, healthy controls.

CONCLUSIONS

- CM-101 demonstrated an anti-inflammatory effect by interfering with migration of monocytes and neutrophils to the damaged biliary area in a PSC animal model.
- CM-101 reduced fibrosis and biliary hyperplasia in a PSC animal model.
- CCL24 induced recruitment of monocytes and neutrophils, which is associated with disease severity in patients with PSC.
- CCL24 is a promising therapeutic target for PSC treatment. CM-101 is currently being tested in a Phase 2 study in PSC patients.

ACKNOWLEDGEMENTS

We would like to thank Dr. Francesca Saffioti, Dr. Stephen Barclay, Dr. Deepak Joshi, Dr. Palak Trivedi, Dr. Matthew Cramp, Dr. George Mells, Dr. Emma Culver, Dr. Ella Veitsman, Prof. Assi Nimer, Prof. Eli Zuckerman, Prof. Haim Shirin, Dr. Yoav Luria, Dr. Maria Carlota Londona Hurtado and all patients for data contribution to the research.

CONTACT INFORMATION

Raanan.Greenman@Chemomab.com