CM-101, a CCL24 neutralizing antibody, showed improvements in inflammatory, fibrotic, and metabolic pathways in patients with NASH: Proteomics analysis of a Phase 2a study

Nov. 10-14, 2023

AASLD The Liver Meeting®

Revital Aricha, Tom Snir, Ilan Vaknin, John Lawler, Adi Mor, Matt Frankel

Chemomab Ltd., Israel and United States

Introduction

CCL24 (eotaxin-2) is a chemokine that promotes cell trafficking and regulates inflammatory and fibrotic processes. CCL24 plays a central role in the development of hepatic fibrosis and liver injury . Patients with non-alcoholic steatohepatitis (NASH) were noted to have elevated levels of CCL24 and CCR3 in liver and blood samples (Segal-Salto M et al.,2020). Phase 2a study (NCT05824156) of CM-101 in NASH patients, met safety objectives and treatment with CM-101 showed improved liver fibrosis biomarkers and physiological assessments.

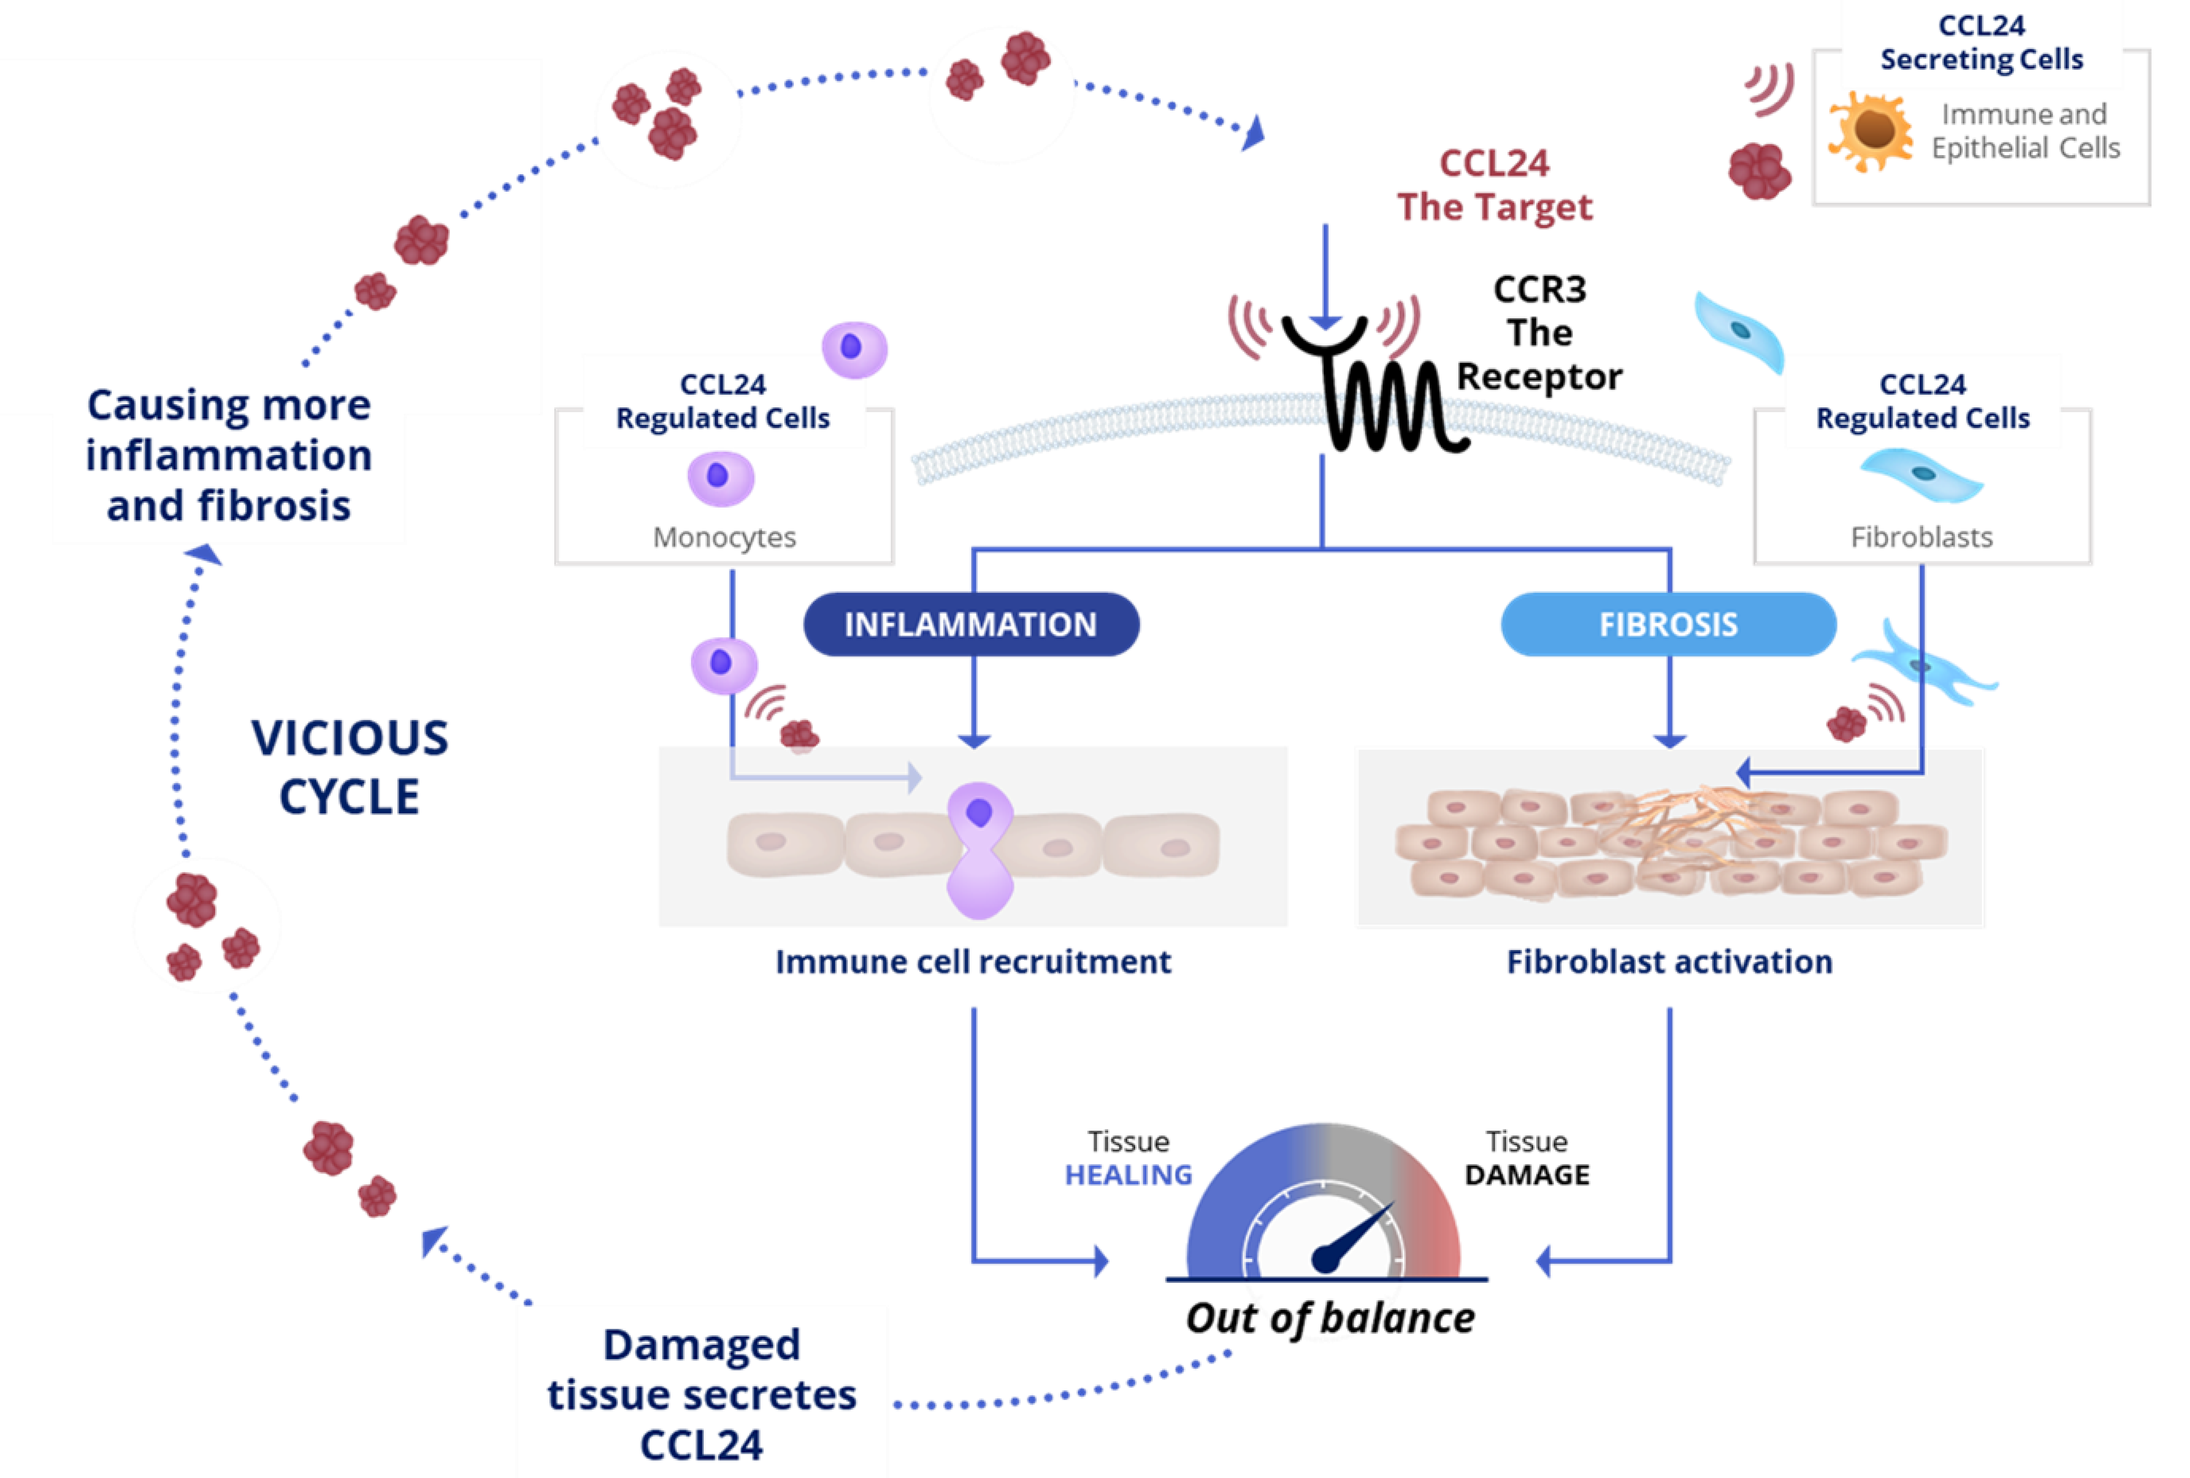
CCL24 Secreting Cells
Immune and Epithelial Cells
CCL24 The Target
CCR3 The Receptor
CCL24 Regulated Cells
Monocytes
CCL24 Regulated Cells
Fibroblasts
Causing more inflammation and fibrosis
VICIOUS CYCLE
INFLAMMATION
FIBROSIS
Immune cell recruitment
Fibroblast activation
Tissue HEALING
Tissue DAMAGE
Out of balance
Damaged tissue secretes CCL24

Aim

We further elucidate the effect of CM-101 on disease related pathways through an analysis of proteomic data from the serum of patients in the phase 2a NASH study.

Method

Twenty-three patients with NASH, diagnosed by liver biopsy, were randomized to receive 8 administrations of either CM-101 5mg/kg (n=14) or placebo (n=9) subcutaneously, every 2 weeks. End of study (EOS) visit occurred 6 weeks post last treatment, at week 20. Sera from patients were analyzed using the Olink® proximity extension assay (PEA) of 3072 proteins. Differentially expressed proteins between CM-101 and Placebo treated patients were identified using a mixed effect model fitted to the data. To identify pathways differentially modulated from baseline to EOS between CM-101 and placebo groups, Gene Set Enrichment Analysis (GSEA) and Ingenuity Pathway analysis (IPA®) were performed.

Results

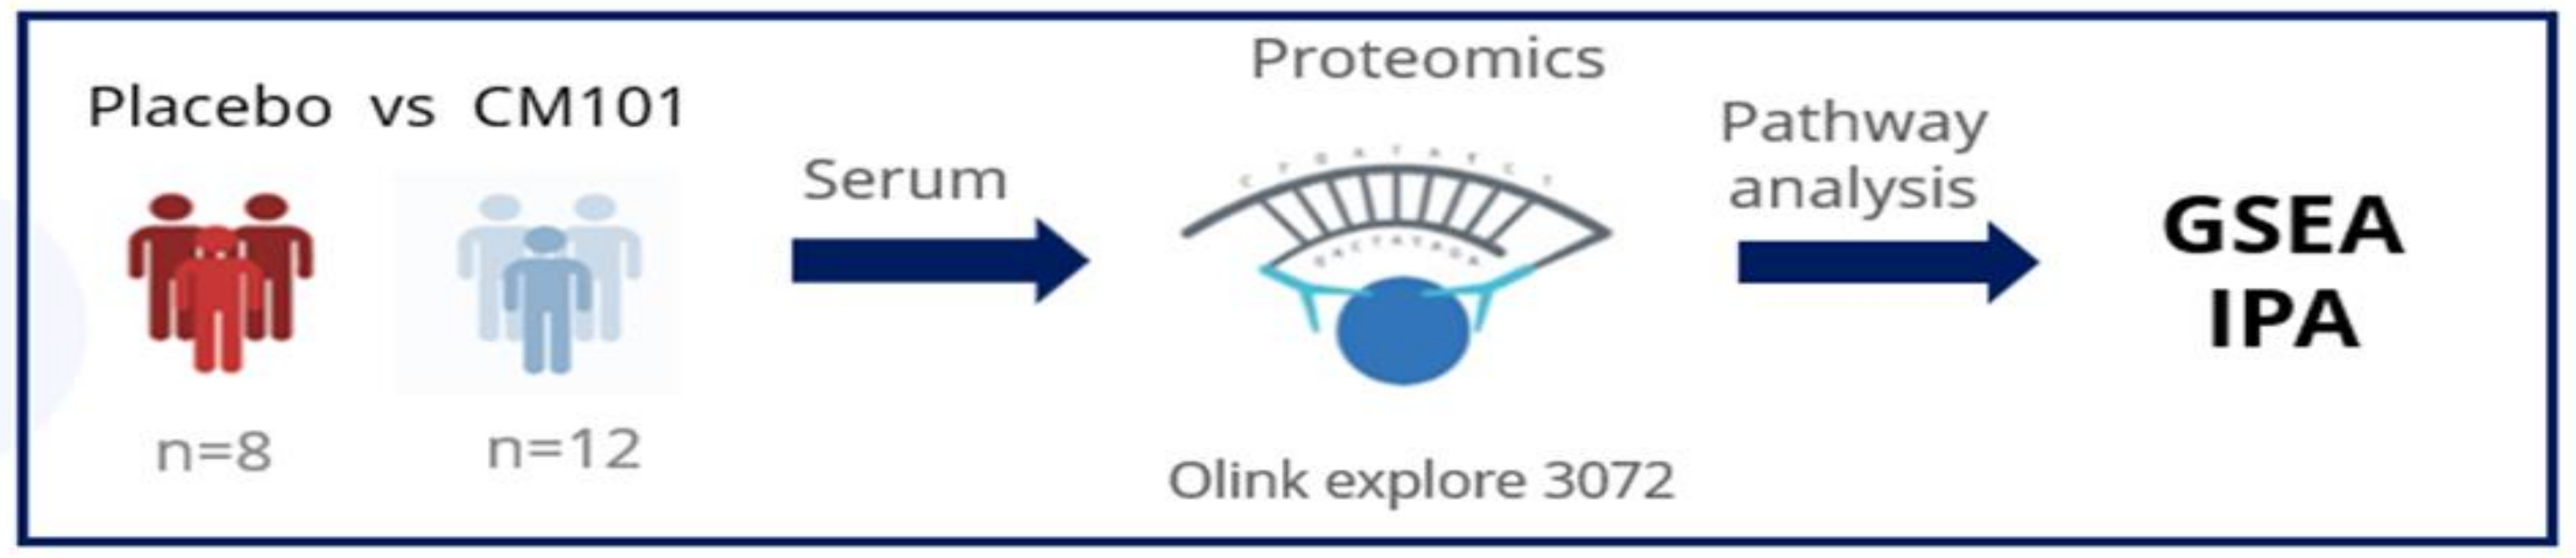
Placebo vs CM101
n=8
n=12
Serum
Proteomics
Olink explore 3072
Pathway analysis
GSEA
IPA

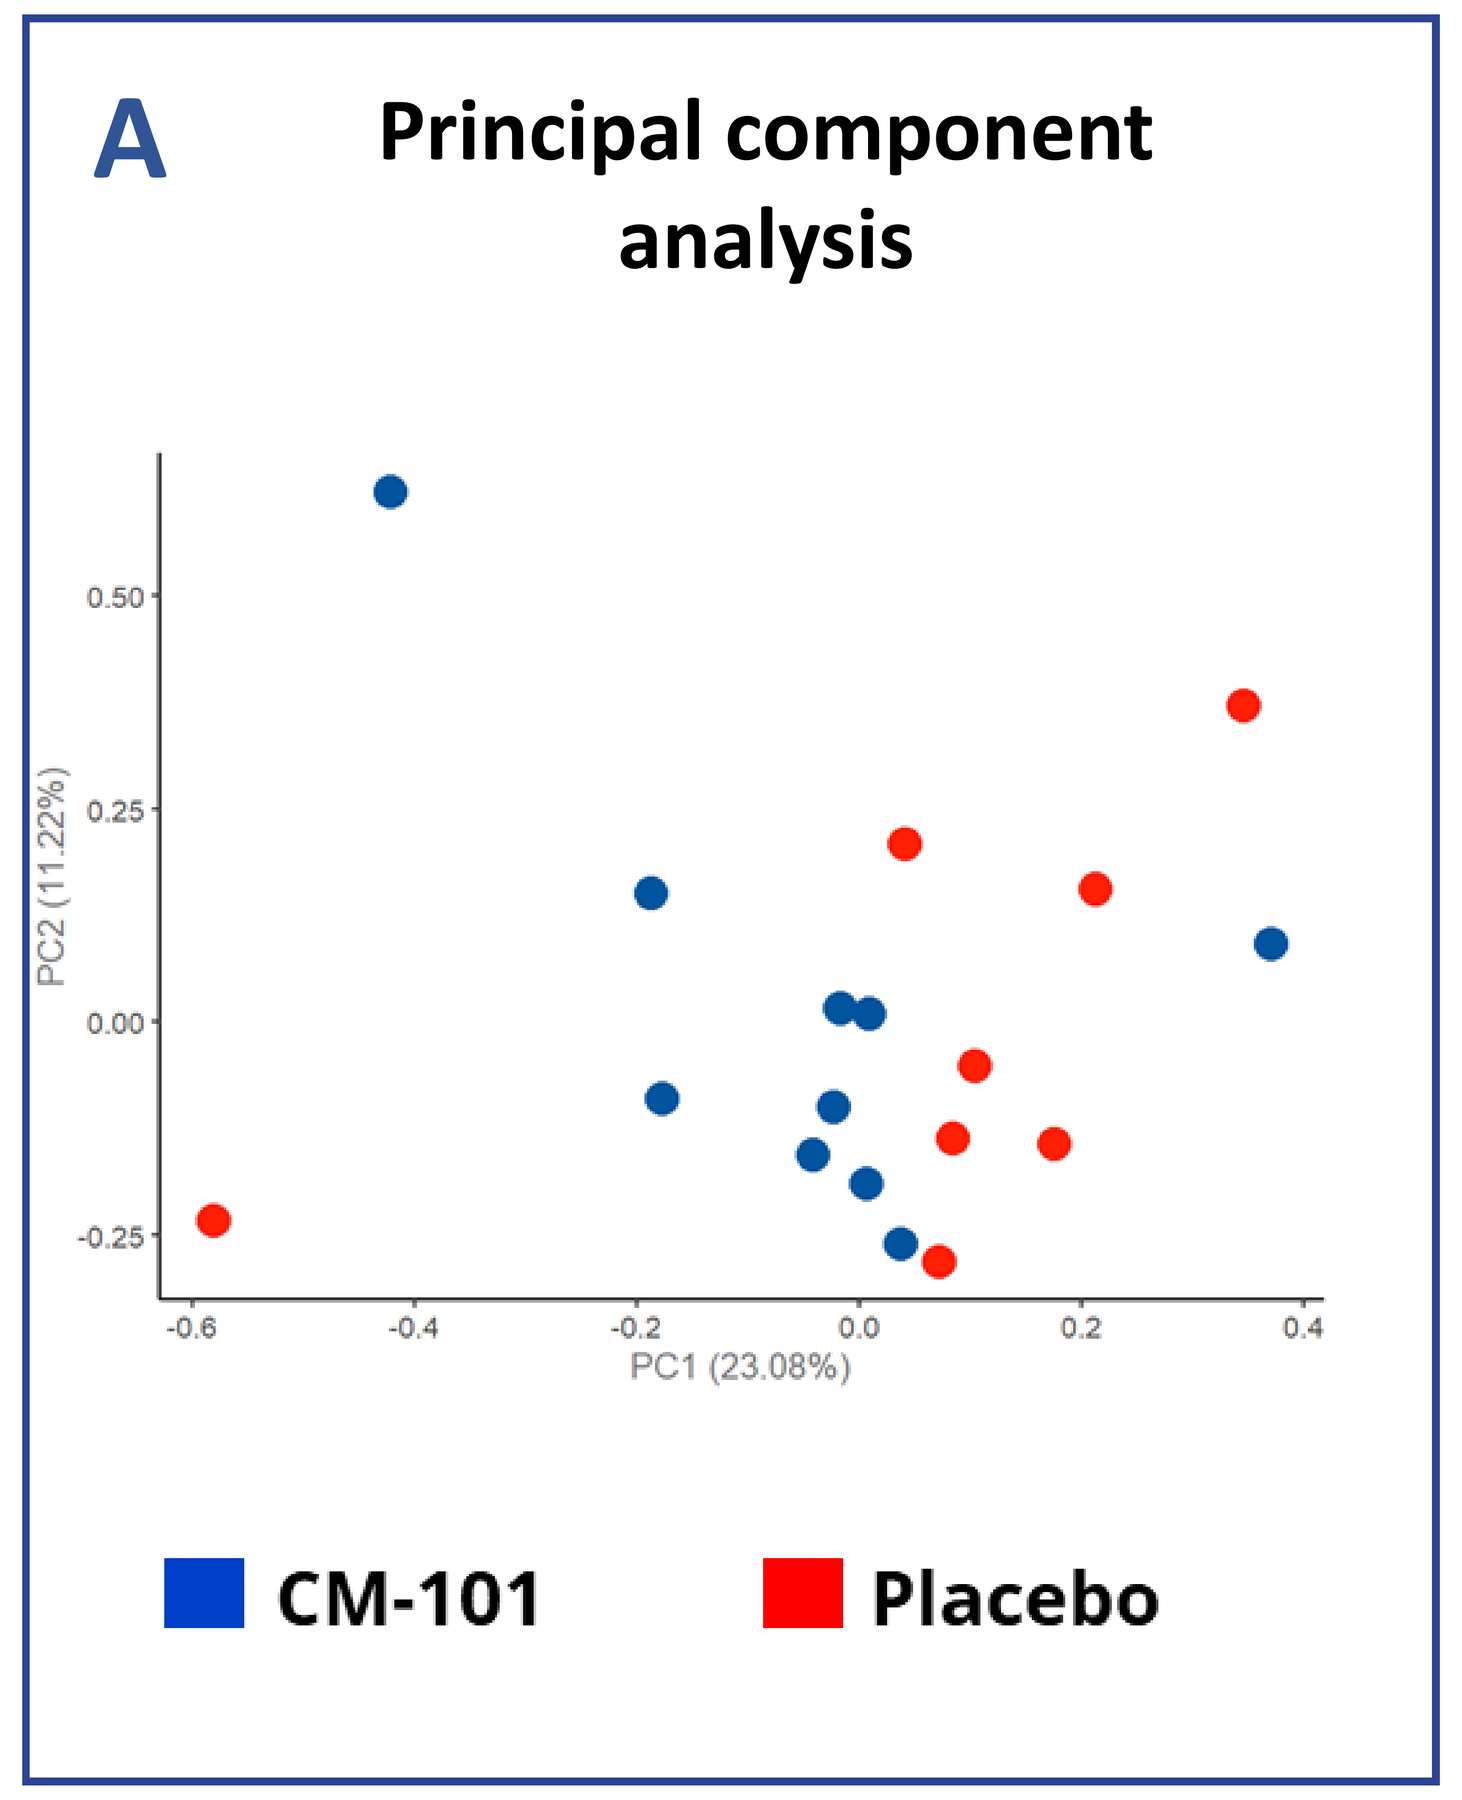
A
Principal component analysis
PC2 (11.22%)
PC1 (23.08%)
CM-101
Placebo

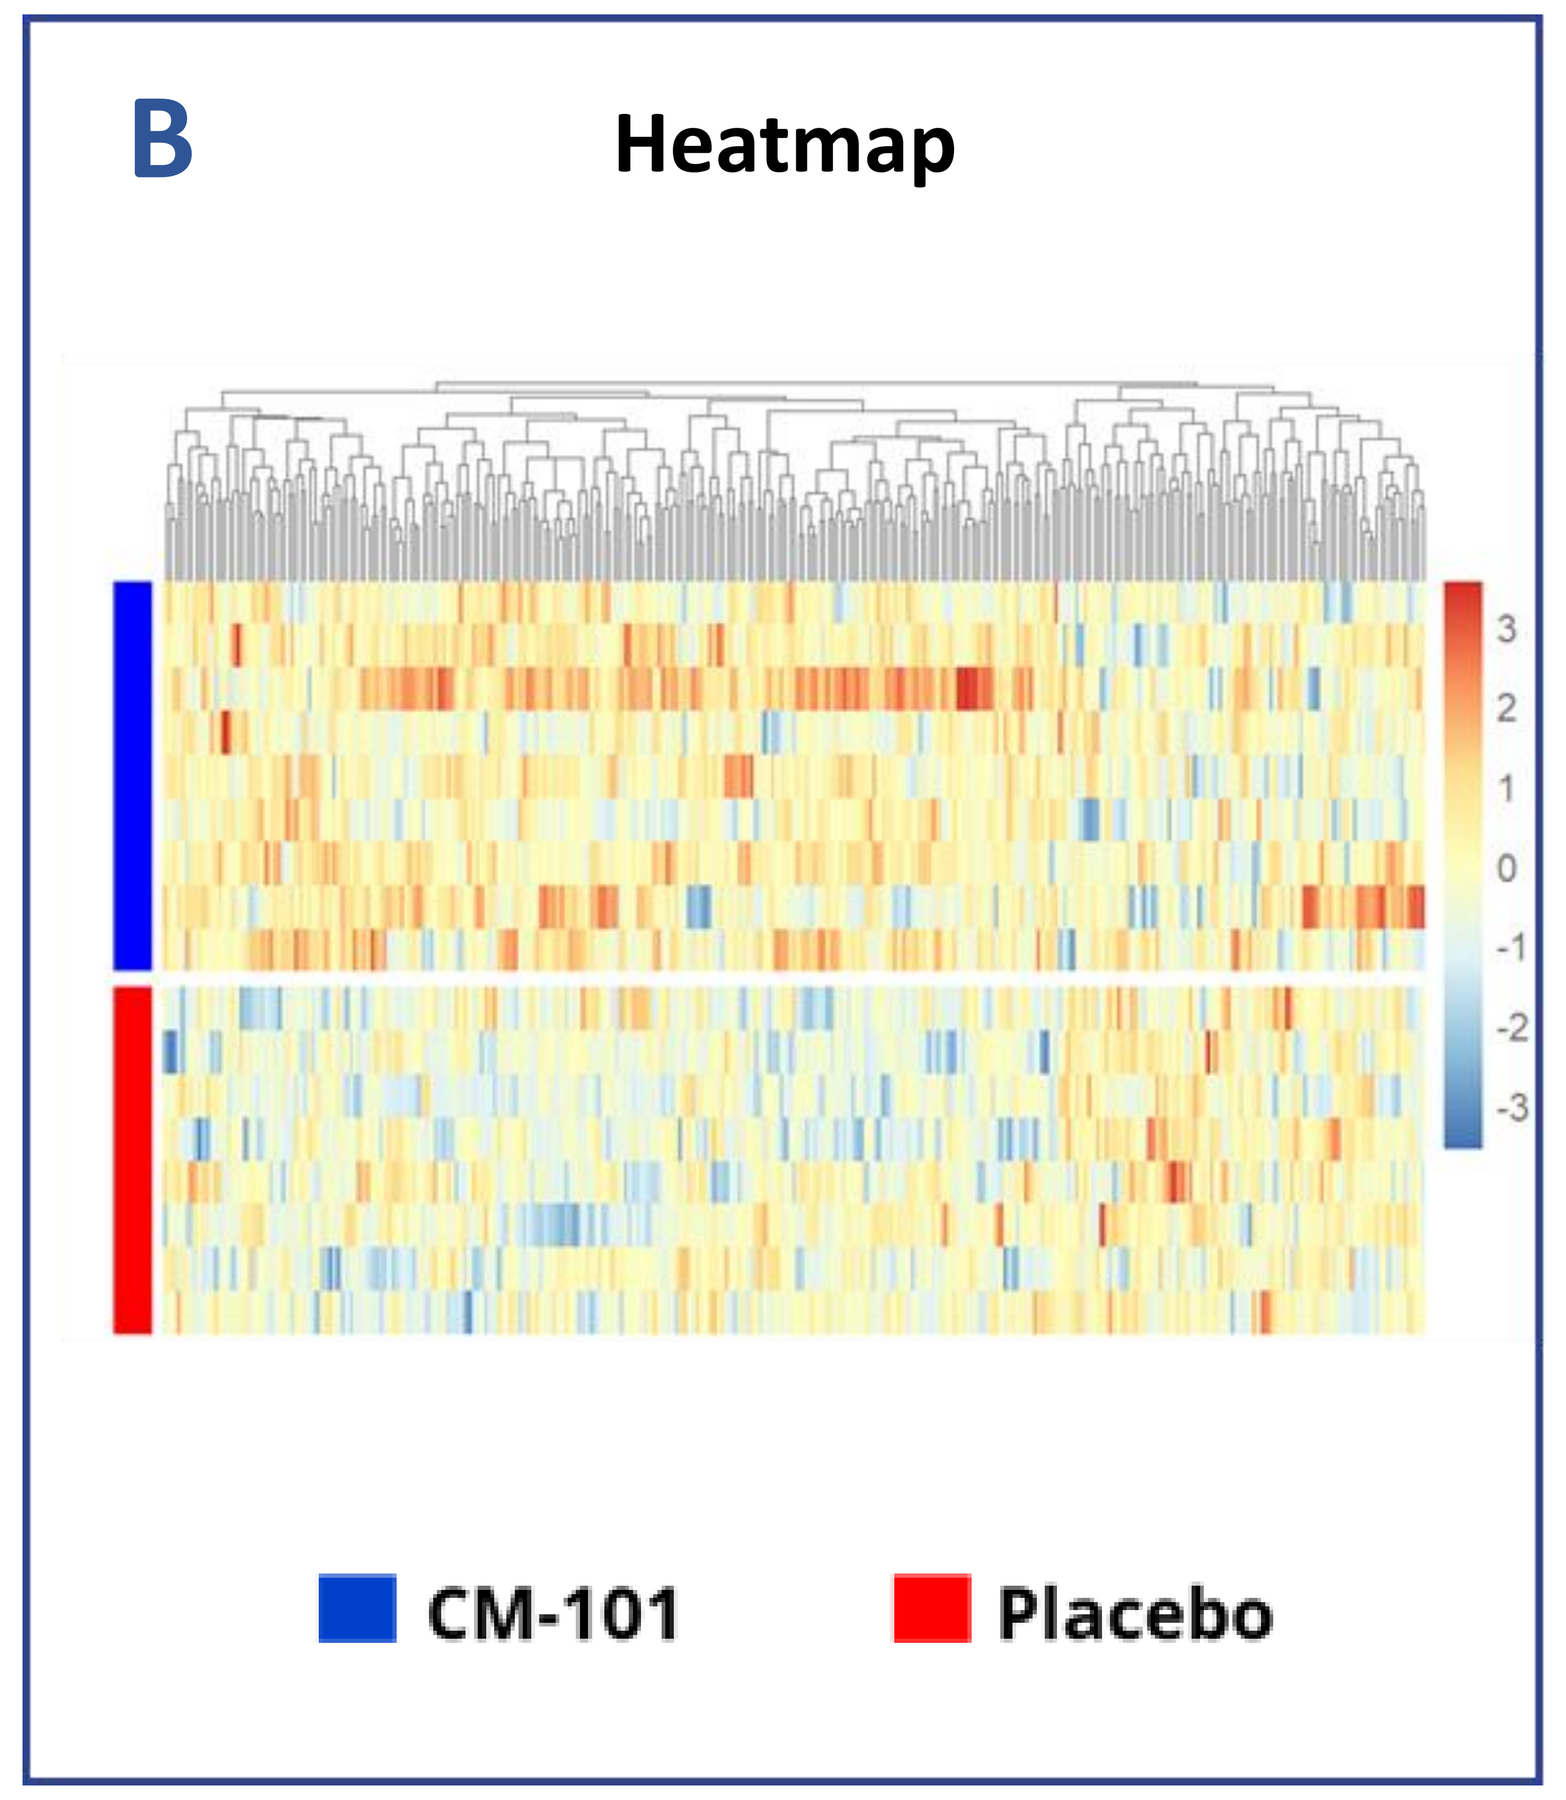
B
Heatmap
CM-101
Placebo

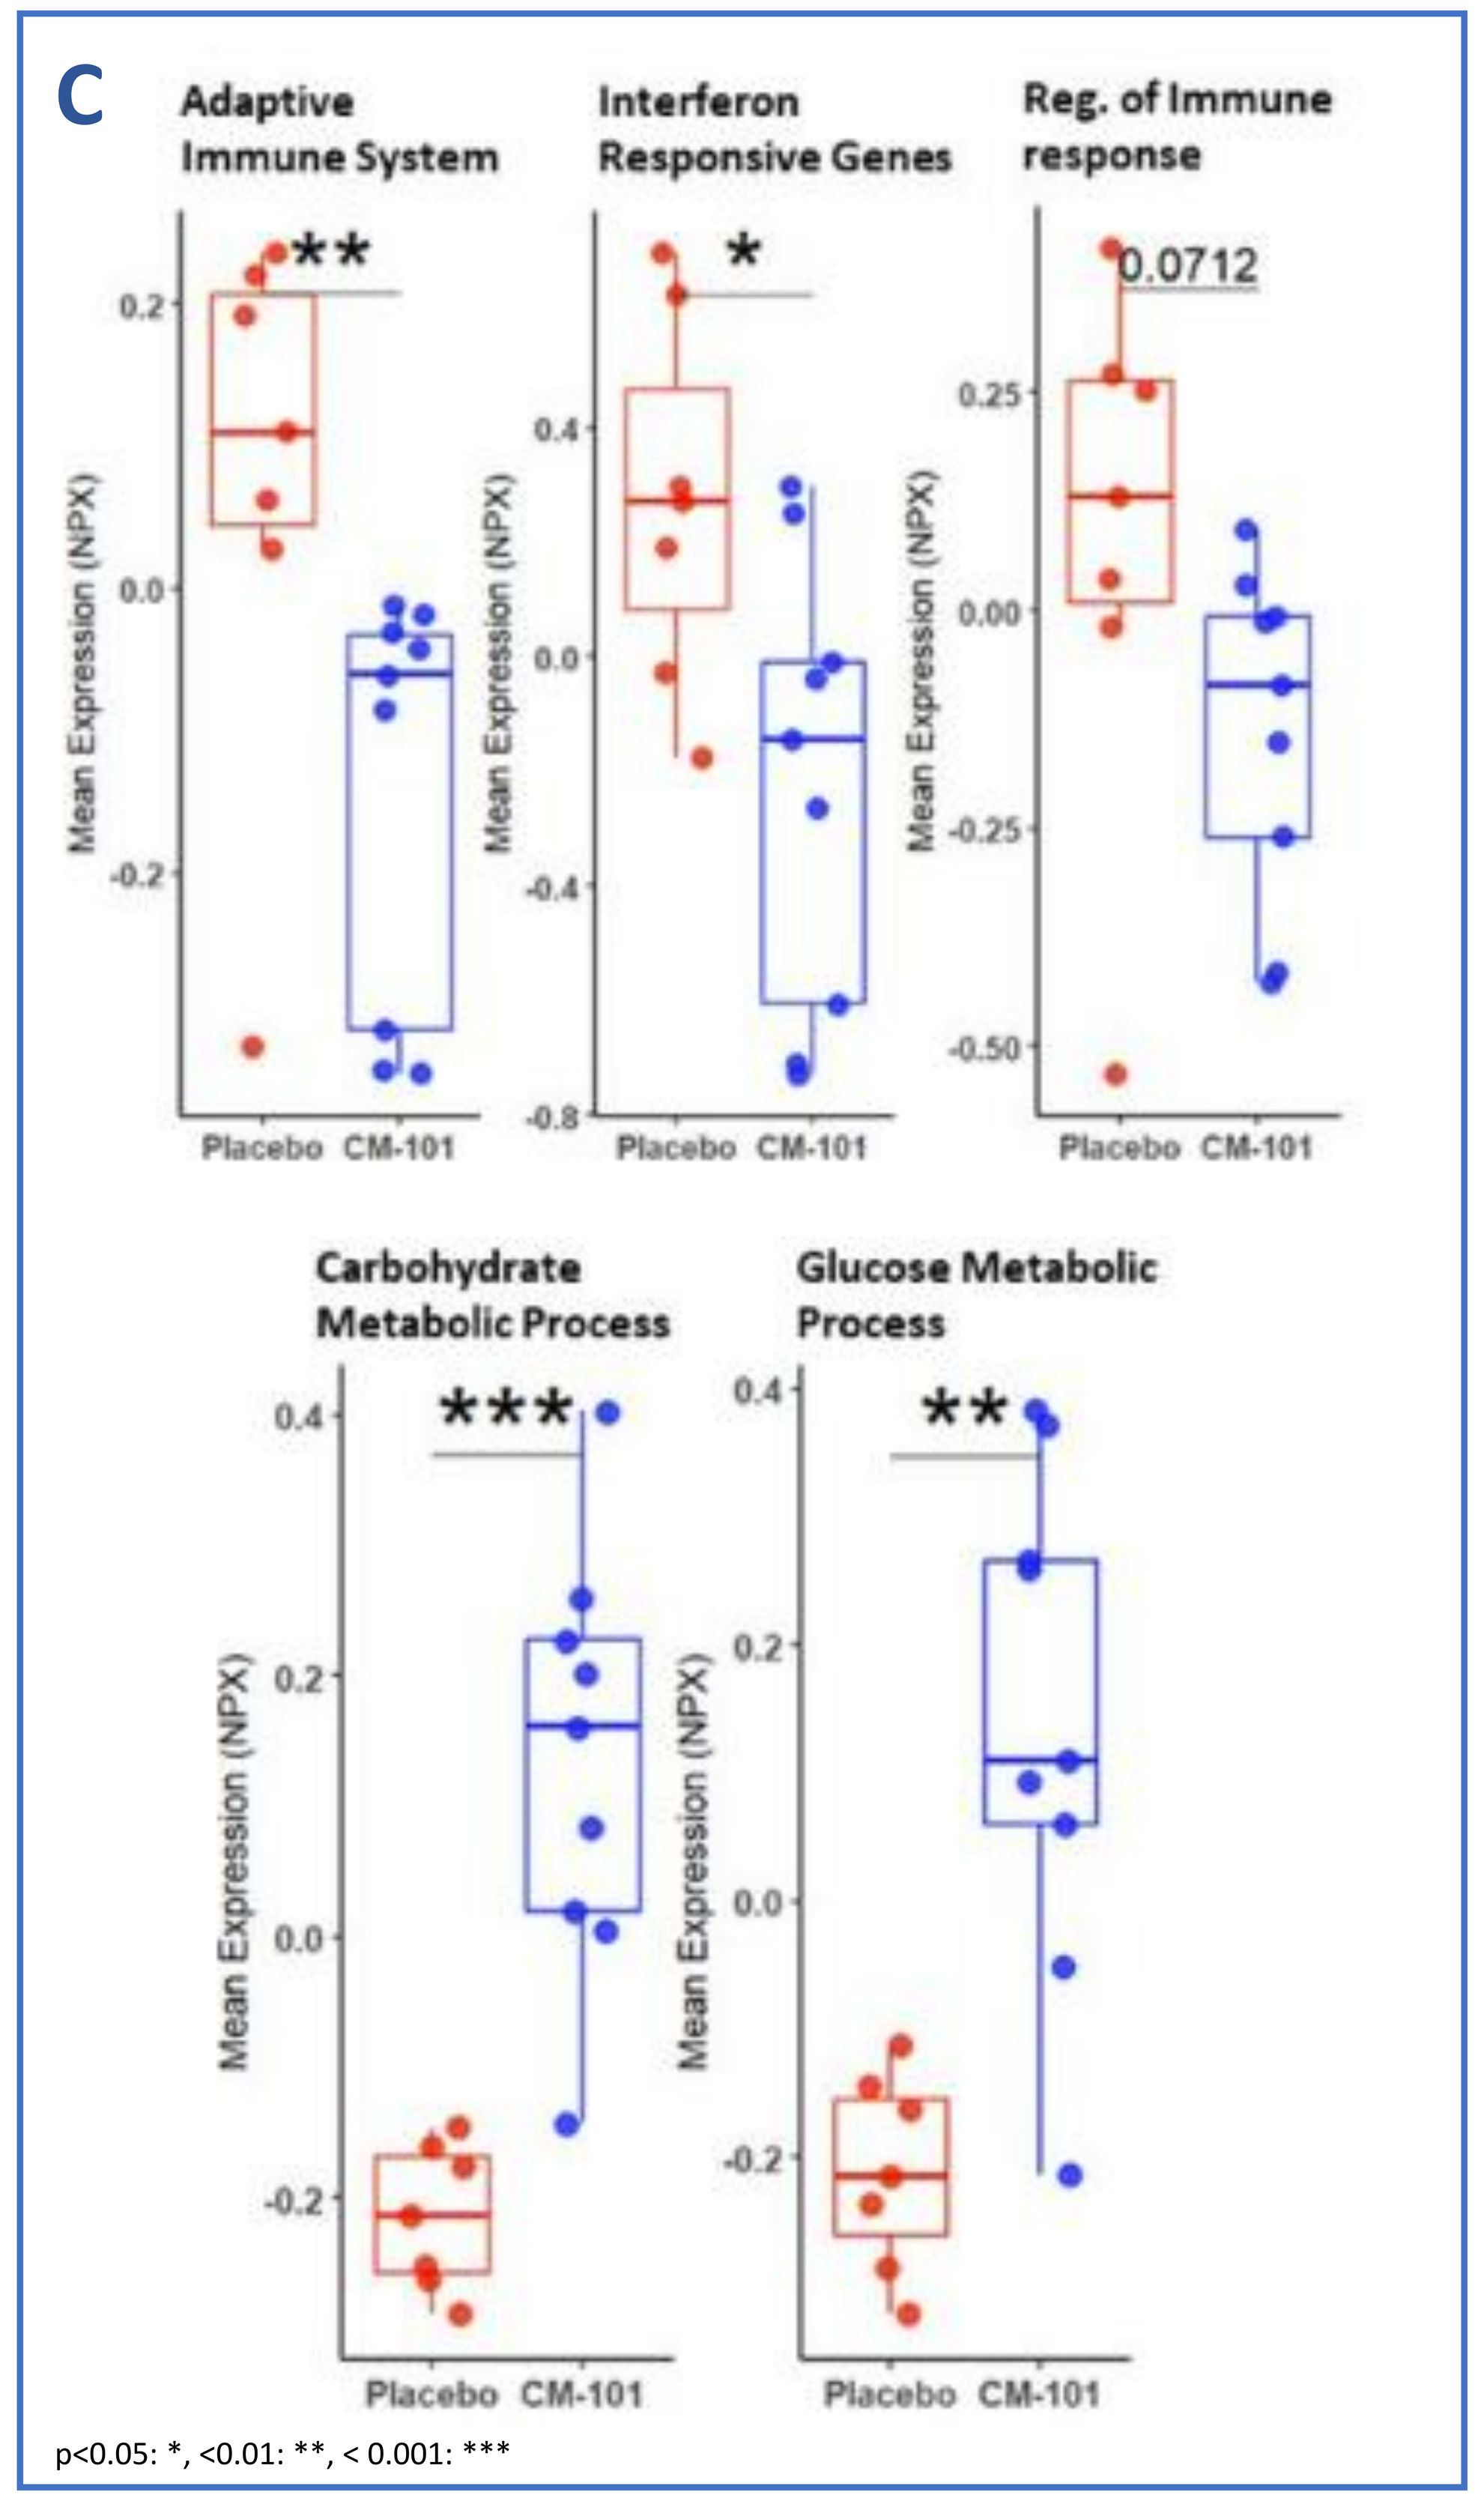
C
Adaptive Immune System
**
Interferon Responsive Genes
*
Reg. of Immune response
0.0712
Carbohydrate Metabolic Process

Glucose Metabolic Process
**
Mean Expression (NPX)
Placebo CM-101
p<0.05: *, <0.01: **, < 0.001: ***

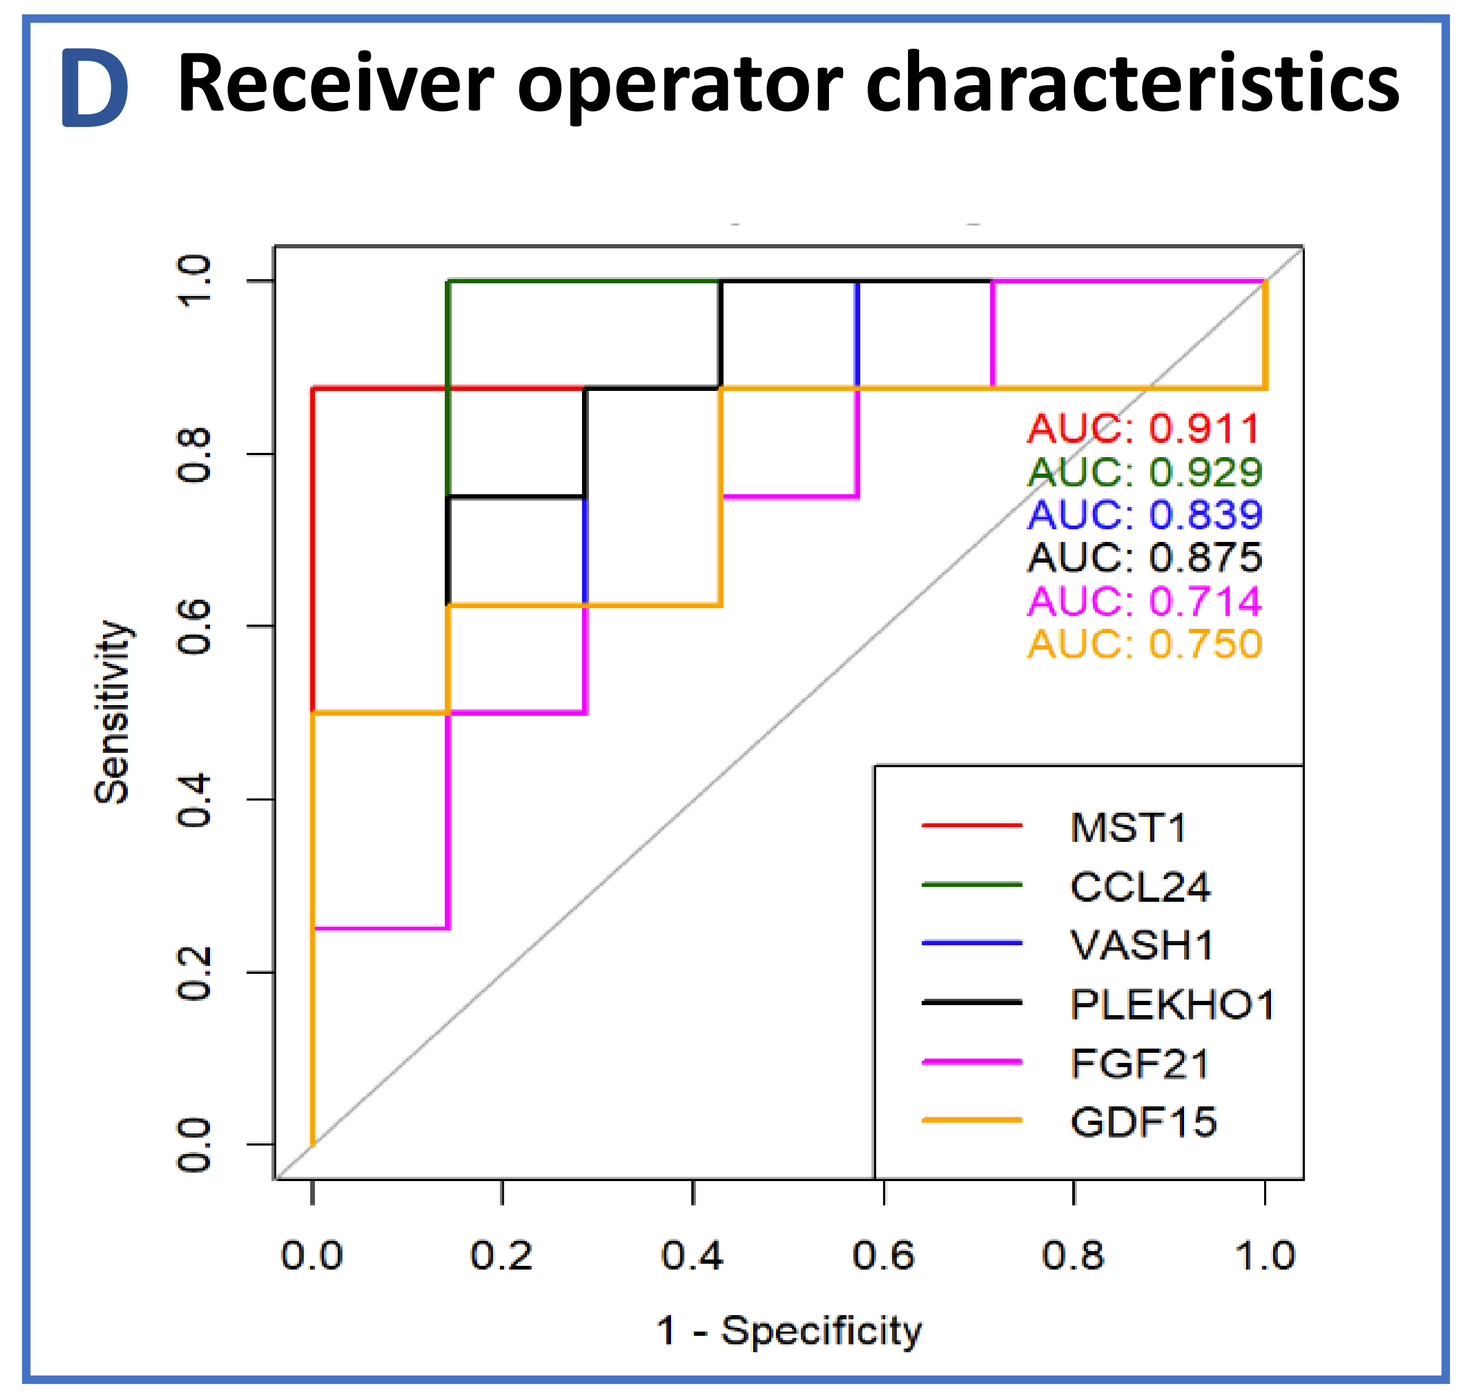
D Receiver operator characteristics
AUC: 0.911
AUC: 0.929
AUC: 0.839
AUC: 0.875
AUC: 0.714
AUC: 0.750
MST1
CCL24
VASH1
PLEKHO1
FGF21
GDF15
Sensitivity
1 - Specificity

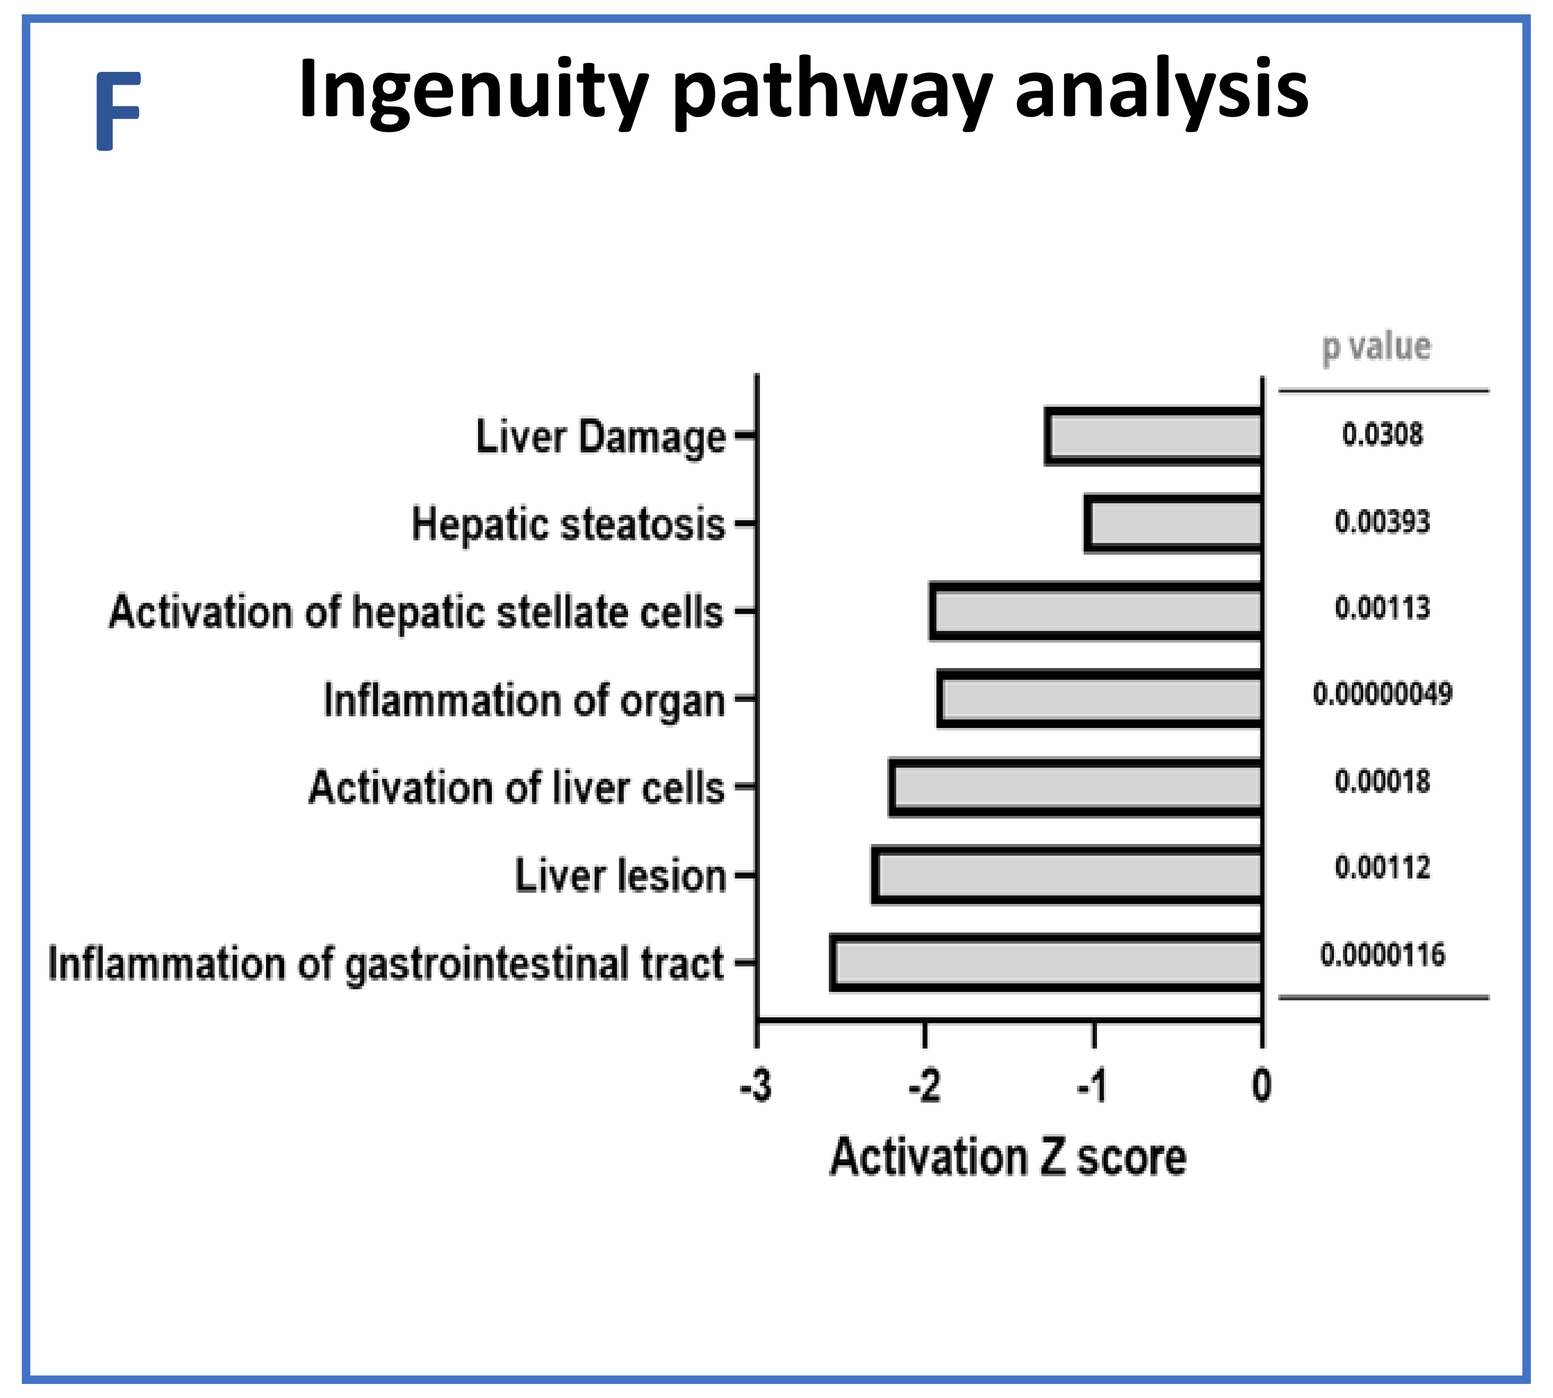
F
Ingenuity pathway analysis
p value
Liver Damage 0.0308
Hepatic steatosis 0.00393
Activation of hepatic stellate cells 0.00113
Inflammation of organ 0.00000049
Activation of liver cells 0.00018
Liver lesion 0.00112
Inflammation of gastrointestinal tract 0.0000116
Activation Z score

Proteome profiling analysis differentiate between CM-101 and Placebo groups

(A) Principal component analysis summarizing the differences in the proteomic profile of each treatment group. (B) Heatmap of significant (p<0.05, by linear mixed model) proteins altered in the treatment group compared to placebo. Values are centered and scaled. (C) Boxplots of key pathways showing mean fold change (NPX values) across all proteins in a given pathway, for each treatment group (p<0.05: *, <0.01: **, < 0.001: ***). (D) Receiver Operator Characteristics (ROC) of selected proteins, showing sensitivity, 1 – specificity and area under the ROC curve. (F) Ingenuity pathway analysis (IPA)- Diseases and Bio Functions and Toxicity Functions, filtered for liver and hepatic related pathways, and their corresponding activation z-score and p values.

Phase 2a Study Design

CM-101 Phase 2a Trial in NASH
Randomized, Double-Blind, Placebo-Controlled

	Total Study Duration 20 weeks	
6-Week Screening	14-Week (8 administrations) Double-blind Treatment Period	6-week Follow-up
N =23 patients 2:1 (Active vs. Placebo)	CM-101 5mg/kg SQ Q2W (n=14)	Post-dosing safety follow-up
	Placebo SQ Q2W (n=9)	

Primary Endpoint:

- Safety and tolerability of CM-101 in subjects with NASH as assessed by adverse events and serious adverse events.

Selected Secondary Endpoints:

- CM-101 serum PK profile over 20 weeks of repeated SC administrations
- Development of anti-drug antibodies (ADA)
- Change from baseline in serum biomarkers for NASH pathogenesis, inflammatory, fibrotic and pharmacodynamic parameters
- Change from baseline in liver stiffness using transient elastography

Conclusion

- CM-101 5mg/kg administered SQ every 2 weeks for a relatively short duration of 14 weeks was safe and well tolerated.
- CM-101 attenuates fibrotic and inflammatory pathways while improving metabolic pathways – all essential in mitigating the sequelae associated with progressive NASH and other fibrotic liver diseases
- Results from this study provided evidence supporting CCL24 as a potential therapeutic target for patients with liver disease driven by inflammation and fibrosis.

Disclosures

TS, RA, JL, AM, IV and MF are employees of Chemomab

CONTACT INFORMATION: matt.frankel@chemomab.com